Poster 34

Phase 3 Study Comparing the Efficacy and Safety of Sustained-Release Granisetron (APF530) and Palonosetron in the Prevention of Chemotherapy-Induced Nausea and Vomiting (CINV) in Cancer Patients Receiving Moderately or Highly Emetogenic Chemotherapy

Carrie Smith,[1] Nashat Gabrail[1]

[1]Gabrail Cancer Center, Canton, OH

BACKGROUND

- CINV is common in patients with cancer receiving chemotherapy, affecting quality of life and adherence to chemotherapy[1-3]
- An important risk factor for CINV is emetogenicity of the chemotherapy regimen; moderately (MEC) and highly (HEC) emetogenic chemotherapies are associated with the highest incidence of CINV[4]
- 5-Hydroxytryptamine type 3 (5-HT_3) antagonists (eg, granisetron) are first-line therapies for prevention of CINV, used to prevent both acute (0-24 h after chemotherapy) and delayed (24-120 h after chemotherapy) CINV[1-3]
- Palonosetron, which has a half-life of ~40 hours, is the preferred 5-HT_3 antagonist according to treatment guidelines[1-3]
- However, patients continue to experience both acute and delayed CINV, and improved approaches to CINV prevention are still needed, particularly in the delayed setting
- APF530 is a novel formulation of granisetron designed to provide slow and sustained release of granisetron
- In clinical studies, a single subcutaneous (SC) dose of APF530 provided sustained therapeutic drug levels for over 5 days (> 120 h)[5,6]
- Nurses provide essential care for patients with cancer experiencing CINV, so it is important that they understand the appropriate use of novel antiemetics such as APF530, including the technology used to develop APF530, APF530 handling and administration techniques, and clinical trial results

APF530: BIOERODIBLE TECHNOLOGY

- APF530 contains 2% granisetron and a bioerodible tri(ethylene glycol) poly(ortho ester) (TEG-POE) polymer (**Figure 1**)

Figure 1. TEG-POE Polymer Allowing for Sustained Release of Granisetron

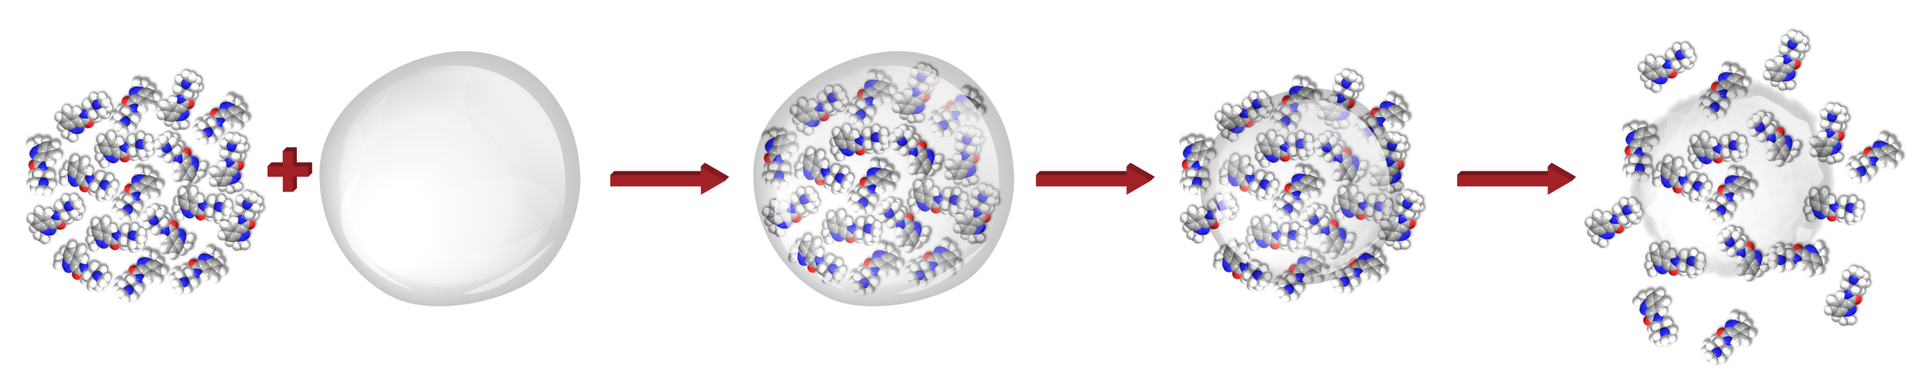

Granisetron
Polymer
Slow, controlled hydrolysis

TEG-POE = tri(ethylene glycol) poly(ortho ester).

- After SC administration, this formulation undergoes slow, controlled hydrolysis, resulting in slow and sustained release of granisetron
- APF530 was designed for the prevention of both acute and delayed CINV associated with MEC and HEC[4,5]

ADMINISTRATION AND HANDLING

- APF530 is provided in a prefilled syringe with a special thin-wall 18-gauge needle (**Figure 2A**), together with a sodium acetate warming pouch (**Figure 2B**)

Figure 2. APF530 Syringe (A) and Sodium Acetate Warming Bag (B)

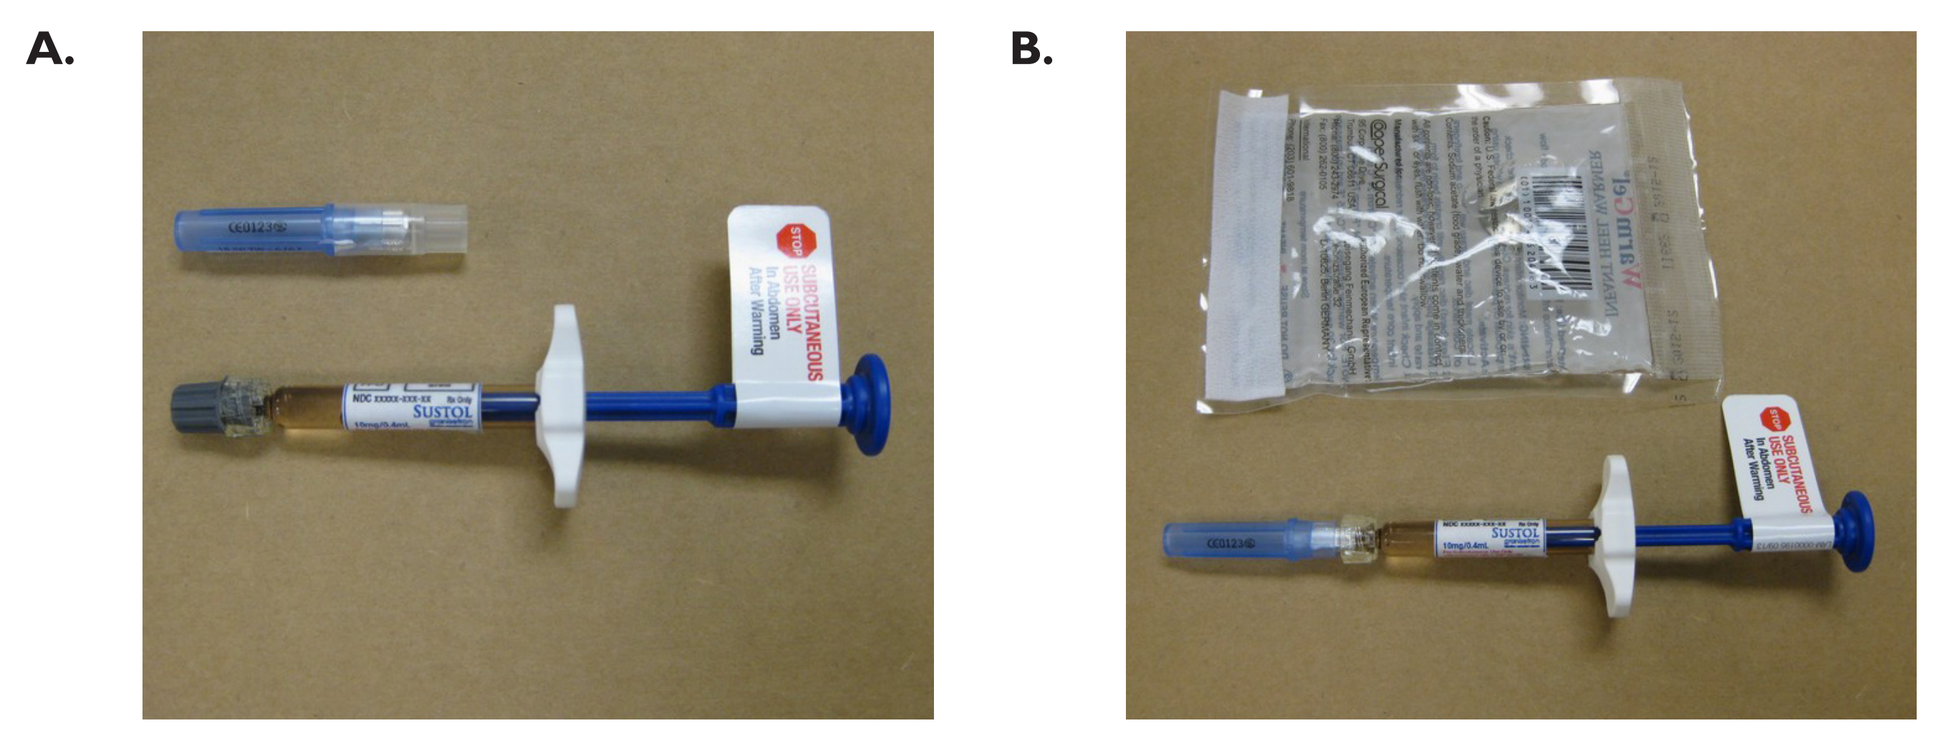

A.
B.

- APF530 is for SC injection only, into the abdomen; a fast-acting local anesthetic is provided and can be used prior to APF530 injection
- The bioerodible polymer used in APF530 is highly viscous, so the force required to inject it is directly proportional to the product's temperature
- APF530 is significantly easier to inject if warmed to body temperature prior to injection
- APF530 should be refrigerated (stored at 40°F or below), then removed 60 minutes prior to use and allowed to reach room temperature
- The prefilled syringe must then be warmed with the sodium acetate warming bag for at least 5 minutes; this will allow APF530 to reach body temperature (**Figure 3A**)
 - The warming bag will stay at the optimal temperature (104°F) for 10 to 15 minutes
 - APF530 can be put back in the refrigerator at any time, and rewarmed several times
 - APF530 can stay unrefrigerated for 7 days, and can be refrigerated up to 2 times
- Once the product has been warmed, and a local anesthetic applied to the abdomen, APF530 should be injected with a slow, firm, and steady push over 20 to 30 seconds (**Figure 3B**)

Figure 3. APF530 Warming (A) and Handling Procedure (B)

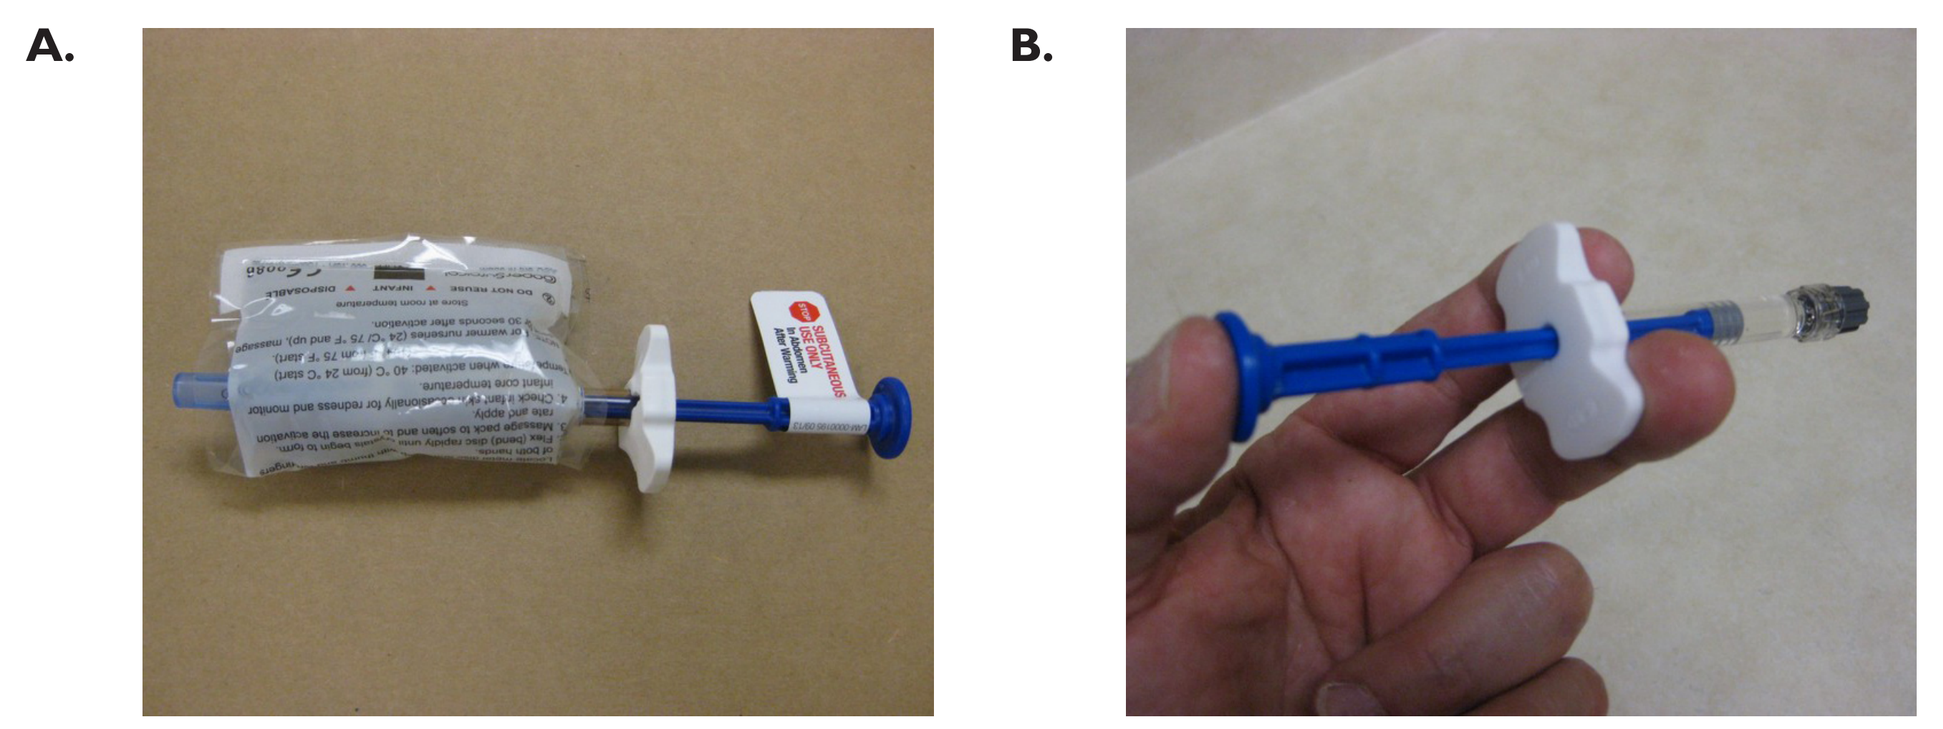

A.
B.

PHASE 3 TRIAL: OBJECTIVES

- The primary objectives of this trial were to establish APF530 noninferiority to palonosetron in preventing acute CINV following MEC or HEC, and delayed CINV following MEC, and to determine APF530 superiority to palonosetron in preventing delayed CINV following HEC
- The primary efficacy endpoint was complete response (CR, using confidence interval [CI] difference for APF530 − palonosetron)

METHODS

- This was a multicenter, randomized, double-blind, double-dummy, parallel-group phase 3 trial
- Chemotherapy-naïve and nonnaïve patients with cancer scheduled to receive single doses of MEC or HEC were randomized to receive APF530 250 or 500 mg SC (granisetron 5 or 10 mg) or palonosetron 0.25 mg intravenously (IV) prior to cycle 1; emetogenicity was defined according to Hesketh et al[4]
- In cycles 2-4, patients who received palonosetron in cycle 1 were randomized to APF530 250 or 500 mg SC; those who received APF530 continued with their cycle 1 APF530 dose (**Figure 4**)
- Standard doses of IV dexamethasone (8 mg IV with MEC, 20 mg IV with HEC) were administered per protocol prior to chemotherapy on day 1; oral dexamethasone (8 mg bid) was given to HEC patients on days 2, 3, and 4
- Treatment cycles were separated by 7 to 28 days

Figure 4. Study Design

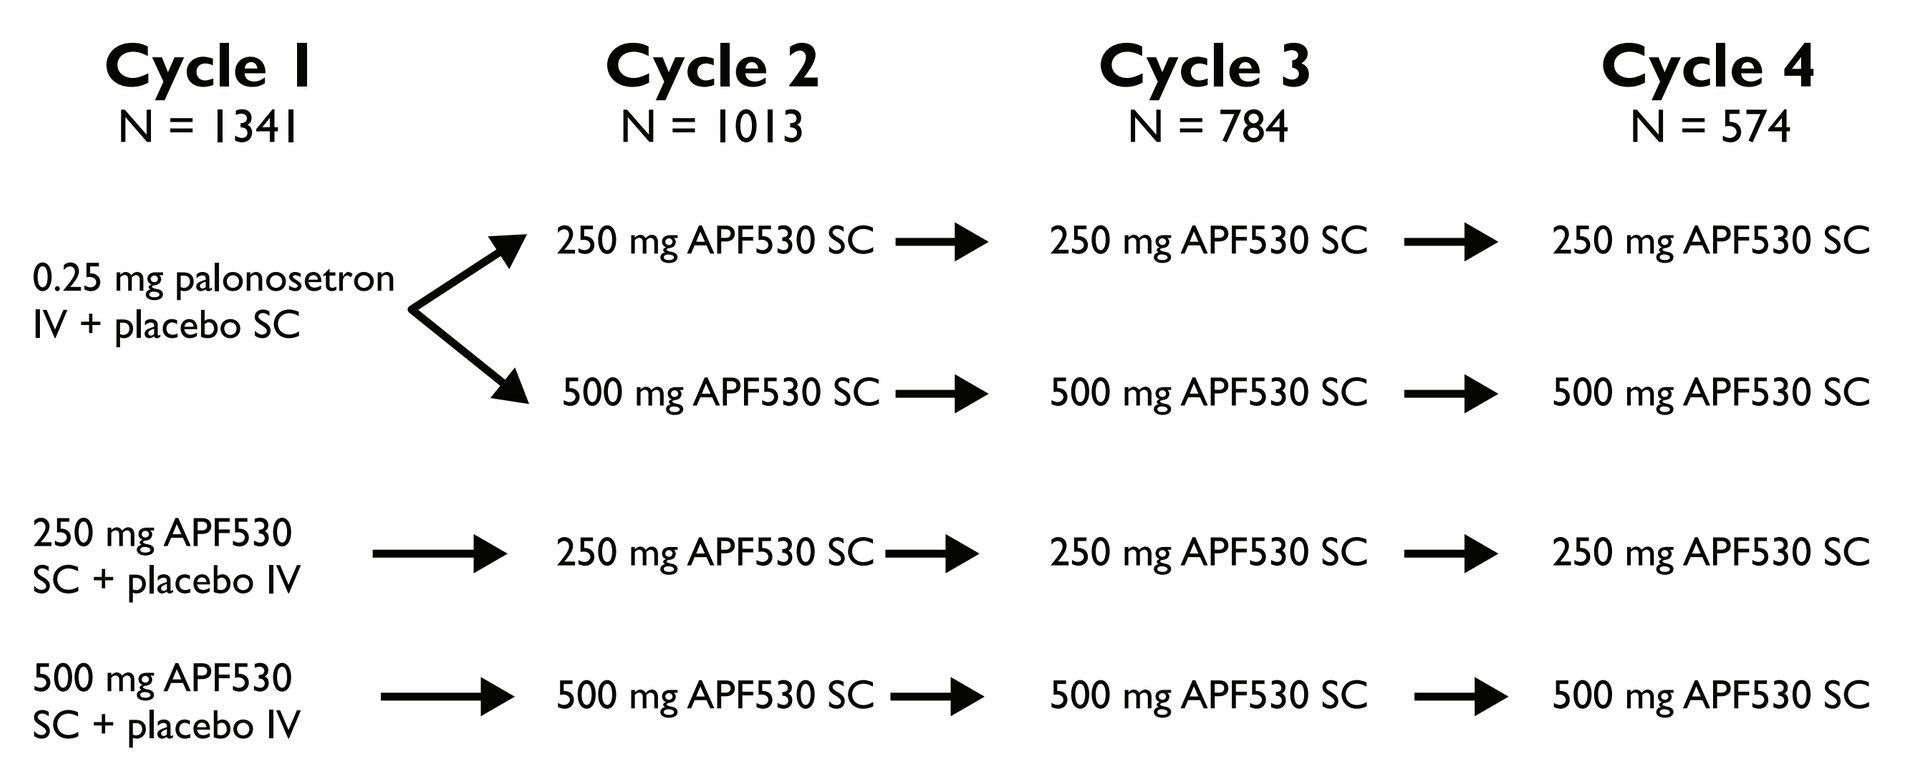

Cycle 1
N = 1341
Cycle 2
N = 1013
Cycle 3
N = 784
Cycle 4
N = 574
0.25 mg palonosetron IV + placebo SC
250 mg APF530 SC
500 mg APF530 SC
250 mg APF530 SC + placebo IV
500 mg APF530 SC + placebo IV

IV = intravenous; SC = subcutaneous.

- Rates of CR (no emetic episodes and no use of rescue medications), complete control (CC; CR with no more than mild nausea), and total response (TR; CR with no nausea) were evaluated across cycles
- After cycle 1, IV palonosetron was discontinued, and all patients who consented were rerandomized to APF530 250 or 500 mg SC for up to 3 more chemotherapy cycles
- Comparisons between groups were made using Fisher's exact test; noninferiority was demonstrated when the lower bound of the CI for CR (APF530 − palonosetron) was greater than −15%

RESULTS

- Baseline patient demographics and clinical characteristics of the 1395 evaluable patients (653 MEC, 742 HEC) are shown in **Table 1**
- Our center enrolled the highest number of patients (n = 88)

Table 1. Patient Demographics and Clinical Characteristics

	MEC			HEC		
	APF530 250 mg n = 220	APF530 500 mg n = 218	Palo 0.25 mg n = 215	APF530 250 mg n = 244	APF530 500 mg n = 250	Palo 0.25 mg n = 248
Age, mean (SD), y	54.8 (12.8)	55.1 (12.8)	57.3 (12.4)	57.4 (13.3)	56.7 (13.3)	58.1 (13.6)
Female, n (%)	193 (87.7)	182 (83.5)	179 (83.3)	164 (67.2)	157 (62.8)	165 (66.5)
ECOG PS 0-1, n (%)	209 (95.0)	214 (98.2)	206 (95.8)	234 (95.9)	239 (95.6)	237 (95.6)
Race/ethnicity						
White or Caucasian	125 (56.8)	123 (56.4)	143 (66.5)	141 (57.8)	156 (62.4)	150 (60.5)
Asian	67 (30.5)	59 (27.1)	48 (22.3)	62 (25.4)	68 (27.2)	59 (23.8)
Other	28 (12.7)	36 (16.5)	24 (11.2)	41 (16.8)	26 (10.4)	39 (15.7)
Hesketh class, n (%)						
1-2	1 (0.5)	2 (0.9)	2 (0.9)	0	0	1 (0.4)
3	27 (12.3)	36 (16.5)	33 (15.3)	1 (0.4)	2 (0.8)	0
4	190 (86.4)	177 (81.2)	177 (82.3)	5 (2.0)	4 (1.6)	1 (0.4)
5	2 (0.9)	3 (1.4)	2 (0.9)	238 (97.5)	244 (97.6)	245 (98.8)
Time since diagnosis, mean (SD), y	n = 213 0.6 (1.7)	n = 212 1.0 (2.1)	n = 207 0.8 (1.8)	n = 236 0.7 (1.8)	n = 241 0.7 (1.7)	n = 235 0.5 (1.0)
Type of cancer, n (%)						
Lung	17 (7.7)	11 (5.0)	15 (7.0)	70 (28.7)	82 (32.8)	63 (25.4)
Breast	153 (69.5)	142 (65.1)	136 (63.3)	66 (27.0)	69 (27.6)	63 (25.4)
Ovarian	17 (7.7)	17 (7.8)	21 (9.8)	34 (13.9)	33 (13.2)	39 (15.7)
Lymphoma	4 (1.8)	5 (2.3)	1 (0.5)	11 (4.5)	11 (4.4)	13 (5.2)
Prior chemotherapy, n (%)	104 (47.3)	109 (50.0)	106 (49.3)	143 (58.6)	145 (58.0)	138 (55.6)

ECOG PS = Eastern Cooperative Oncology Group performance status; HEC = highly emetogenic chemotherapy; MEC = moderately emetogenic chemotherapy; Palo = palonosetron; SD = standard deviation.

- Most patients had received prior chemotherapy; the most common prior regimens were cyclophosphamide + anthracycline-based in patients receiving MEC (56%), and carboplatin + taxane-based in patients receiving HEC (40%)

Efficacy

- APF530 250 mg and 500 mg were noninferior to palonosetron in preventing acute CINV after MEC and HEC regimens in cycle 1 (**Figure 5A**)
- APF530 500 mg was noninferior to palonosetron in preventing delayed CINV after MEC in cycle 1 (**Figure 5A**)
- APF530 500 mg was not superior to palonosetron in preventing delayed CINV after HEC in cycle 1 (**Figure 5B**)
- There were no significant differences in within-cycle CR rates between APF530 doses during acute and delayed phases for MEC and HEC in cycle 1 (**Figure 5A, B**) or in later cycles

Figure 5. CR Rates With APF530 for Patients With Cancer Treated With MEC (A) and HEC (B) Regimens Over 4 Cycles[7,8]

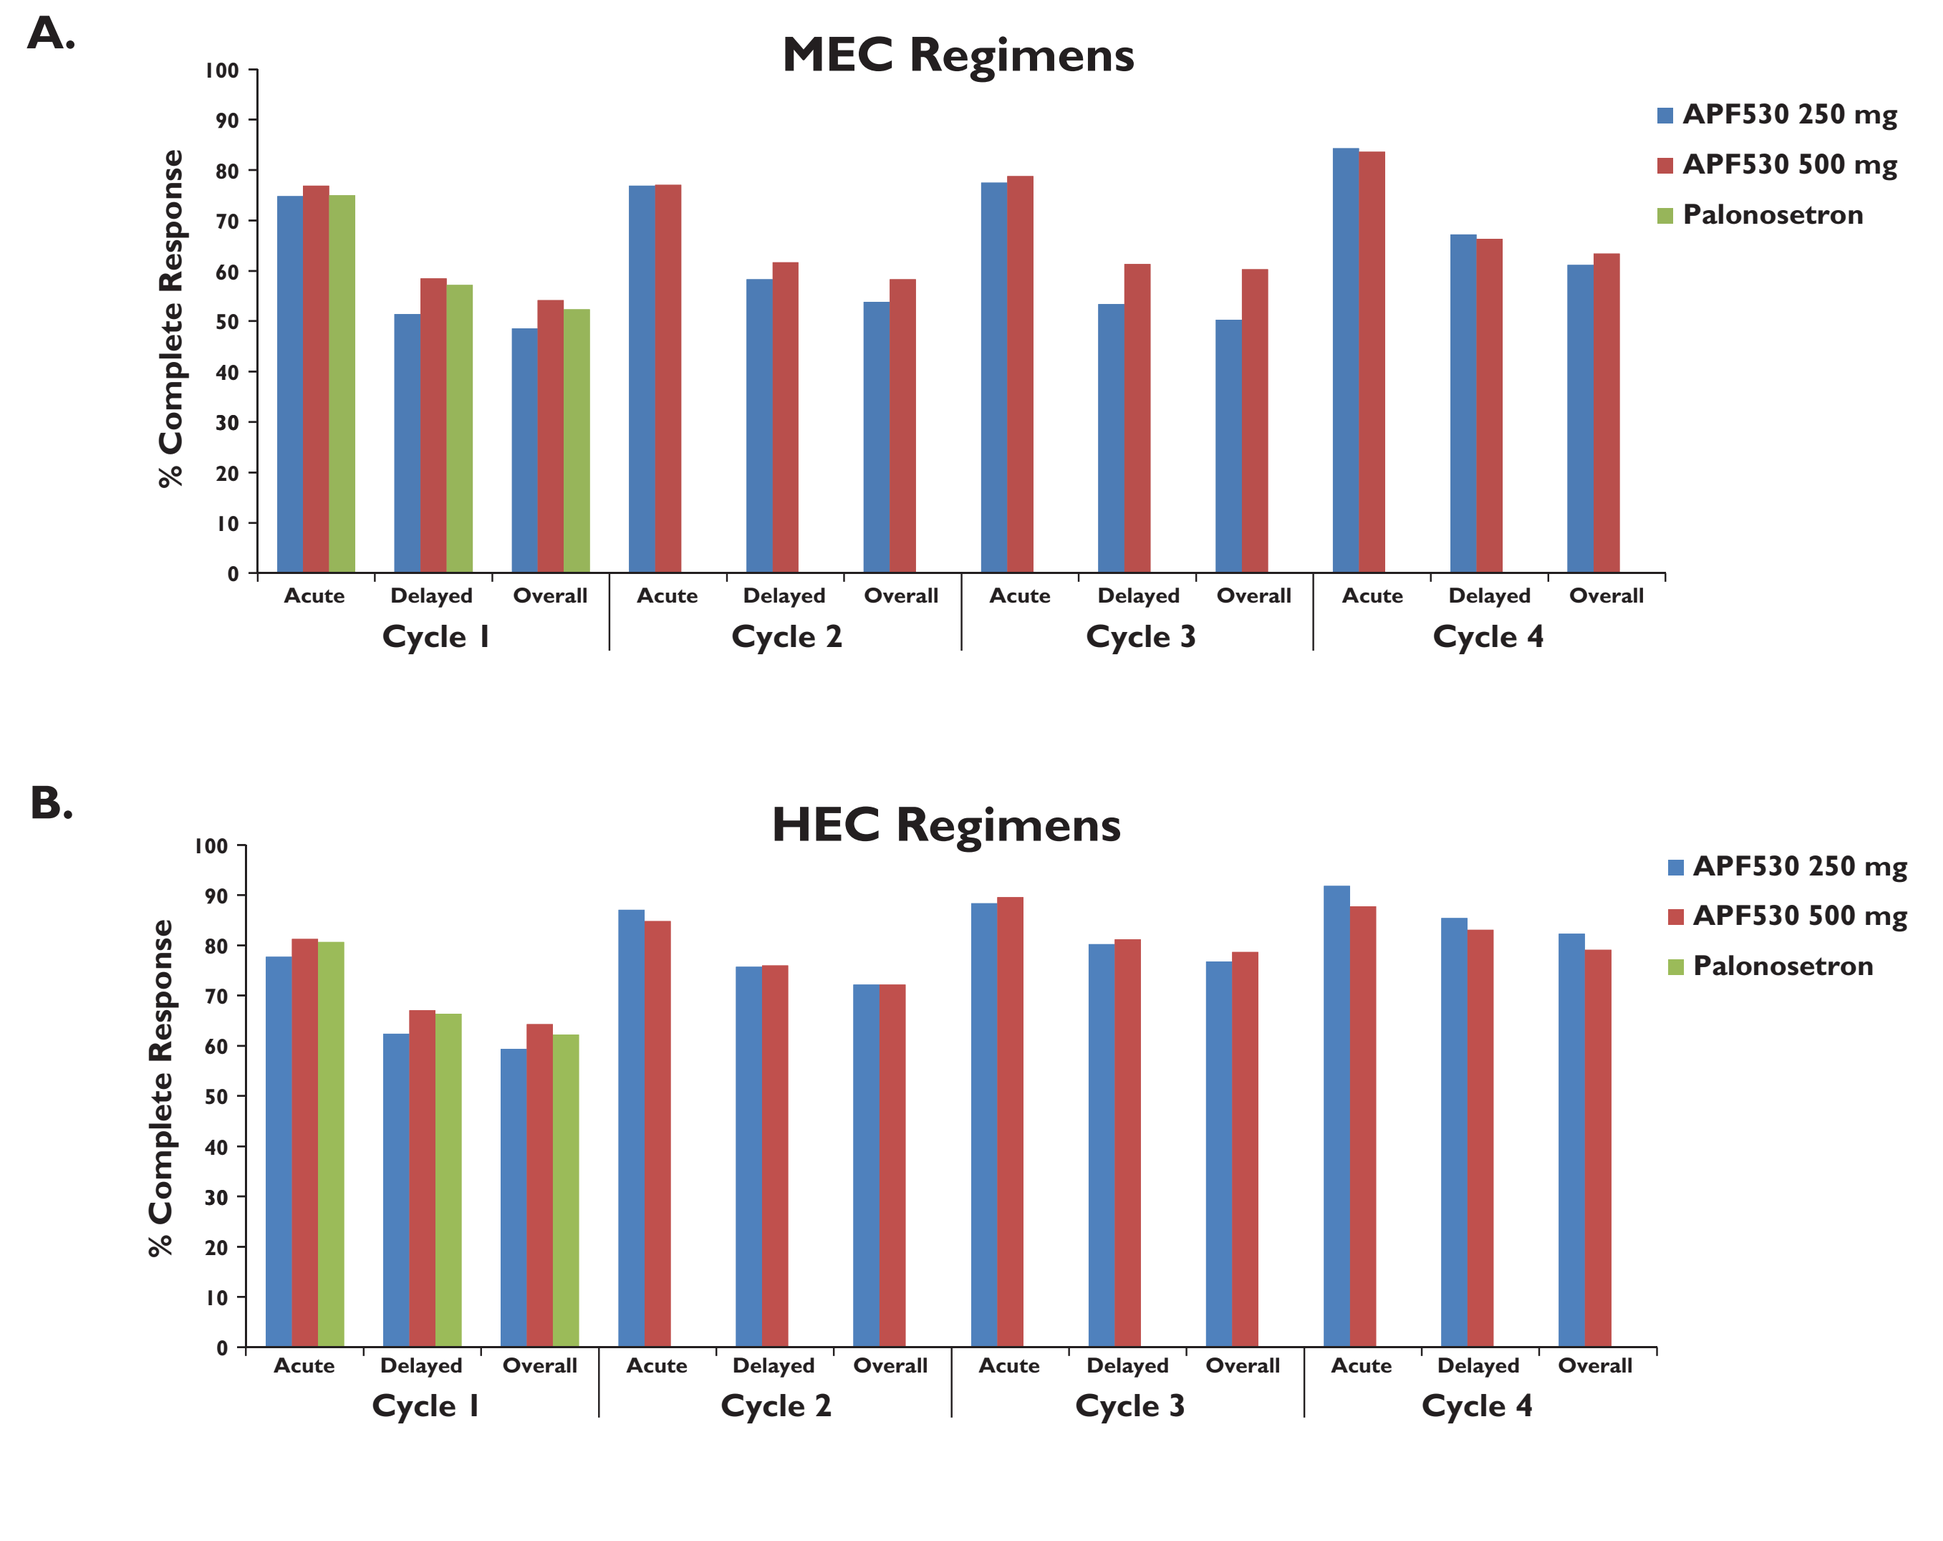

A.
MEC Regimens
% Complete Response
0
10
20
30
40
50
60
70
80
90
100
APF530 250 mg
APF530 500 mg
Palonosetron
Acute
Delayed
Overall
Cycle 1
Cycle 2
Cycle 3
Cycle 4
B.
HEC Regimens

HEC = highly emetogenic chemotherapy; MEC = moderately emetogenic chemotherapy.

- In cycle 1, ≥ 75% of patients receiving APF530 had a CR during the acute phase of CINV and ≥ 50% had a CR during the delayed phase of CINV, with somewhat higher rates among patients receiving HEC than those receiving MEC; this pattern was maintained across cycles 2 to 4[9]
- There was no significant difference between APF530 and palonosetron in overall CR rate of chemotherapy-naïve (58% vs 55%) and nonnaïve (63% vs 55%) patients[8]
- In a pharmacokinetic analysis of APF530, concentrations of granisetron were detected in the plasma for 120 hours during cycle 1 (**Figure 6**)

Figure 6. Plasma Concentrations of Granisetron After APF530 Administration

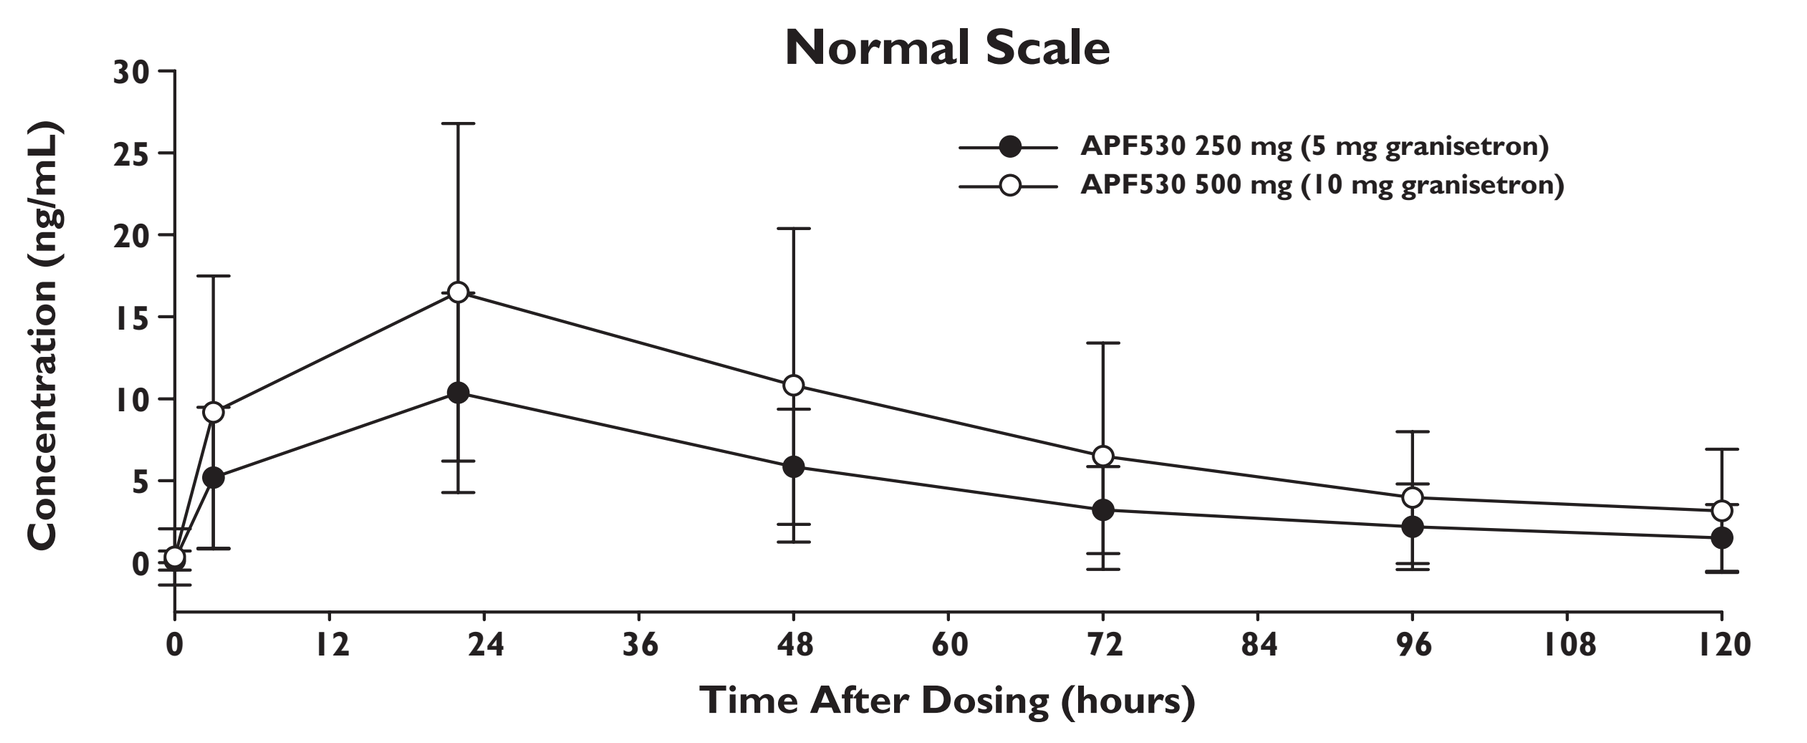

Normal Scale
APF530 250 mg (5 mg granisetron)
APF530 500 mg (10 mg granisetron)
Concentration (ng/mL)
0
5
10
15
20
25
30
0
12
24
36
48
60
72
84
96
108
120
Time After Dosing (hours)

Safety

- Adverse events (AEs) were as anticipated for granisetron, and the frequency of AEs was similar across all treatment groups in cycle 1
 - AEs were predominantly mild and considered unrelated to treatment
 - Excluding injection-site reactions, the most common AEs across all groups were constipation (13.4%-15.6%), fatigue (11.9%-14%), and nausea (8.9%-12.8%)
 - Injection-site reactions occurred in all treatment groups and most commonly during cycle 1; the most common were bruising (9.1%-19.9%), erythema (3.5%-10.9%), and nodules (0.6%-10.7%)
 - There were 4 deaths in cycle 1 (2 in the APF530 500-mg group, 1 in the APF530 250-mg group, and 1 in the palonosetron group); none was related to treatment
- The frequencies of treatment-related AEs (TRAEs) were also similar between groups (**Table 2**)
 - In cycle 1, 2 patients discontinued because of a TRAE (dyspnea in 1 patient receiving APF530 250 mg and hypersensitivity in 1 patient receiving APF530 500 mg)
 - Treatment-related injection-site reactions were generally mild and resolved over time

Table 2. Treatment-Related Adverse Events (in ≥ 5% of Patients) in Cycle 1

	APF530 250 mg n = 464	APF530 500 mg n = 468	Palo 0.25 mg n = 463
Preferred Term,* n (%)			
Asthenia	3 (0.6)	0	3 (0.6)
Constipation	20 (4.3)	21 (4.5)	14 (3.0)
Diarrhea	6 (1.3)	5 (1.1)	5 (1.1)
Fatigue	6 (1.3)	5 (1.1)	3 (0.6)
Headache	13 (2.8)	13 (2.8)	9 (1.9)
Insomnia	3 (0.6)	0	0
Nausea	3 (0.6)	4 (0.9)	3 (0.6)
Injection-site reactions, n (%)			
Bruising	60 (12.9)	73 (15.6)	30 (6.5)
Erythema	28 (6.0)	38 (8.1)	13 (2.8)
Nodules	19 (4.1)	44 (9.4)	3 (0.6)
Pain	14 (3.0)	27 (5.8)	6 (1.3)

*A patient with > 1 event in a given preferred term was counted once within each relatedness category in that preferred term.

CONCLUSIONS

- APF530 is a SC formulation of granisetron in a bioerodible polymer that provides slow and sustained release of granisetron over > 120 hours
- Administration of APF530 SC required no specialized nursing skills
- APF530 was noninferior to palonosetron in preventing acute CINV after MEC and HEC, and noninferior to palonosetron in preventing delayed CINV after MEC
- CR rates with APF530 were sustained over up to 4 cycles of chemotherapy
- APF530 was equally effective in chemotherapy-naïve and nonnaïve patients
- In our experience, APF530 was effective regardless of patient age, sex, tumor type, and prior chemotherapy, important considerations for nurses caring for patients with cancer
- Safety profiles of APF530 and palonosetron were similar, requiring no specialized nursing intervention
- A single SC injection of APF530 may be a convenient alternative to palonosetron for preventing CINV after MEC or HEC, especially in an outpatient setting

REFERENCES

1. Roila et al. *Ann Oncol.* 2010;21(suppl 5):v232-243.
2. Basch et al. *J Clin Oncol.* 2011;29:4189-4198.
3. NCCN Clinical Practice Guidelines in Oncology: Antiemesis Guidelines—v1.2013.
4. Hesketh et al. *J Clin Oncol.* 1997;15:103-109.
5. Barr et al. ASCO 2013. Abstract e20587.
6. Gabrail et al. ASCO 2013. Abstract e20518.
7. Charu et al. MASCC 2012. Abstract 1109.
8. Grous et al. ASCO 2009. Abstract 9627.
9. Boccia et al. ASCO 2013. Abstract 9626.

Copies of this poster obtained through Quick Response Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.