Patient Satisfaction With Control of Emesis Following Chemotherapy:

Comparison of APF530, a Subcutaneous Extended-Release Formulation of Granisetron, Versus Intravenous Palonosetron

Veena Charu,[1] Rebecca Clark-Snow,[2] Nashat Gabrail,[3] Ronald Yanagihara,[4] Martin J. Rosenberg,[5] Erin O'Boyle,[6] John Barr,[6] [1]Pacific Cancer Medical Center, Inc., Anaheim, CA; [2]University of Kansas Cancer Center, Westwood, KS; [3]Gabrail Cancer Center, Canton, OH; [4]Ronald Yanagihara, MD, Gilroy, CA; [5]MAJARO InfoSystems, Santa Clara, CA; [6]A.P. Pharma, Redwood City, CA, USA

BACKGROUND

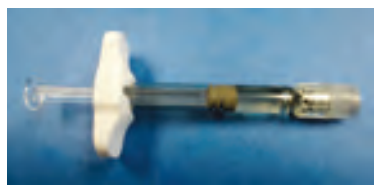

- Prevention and control of acute and delayed chemotherapy-induced nausea and vomiting (CINV) are cornerstones of supportive care for patients with cancer.
- Although several treatment options exist, there is a need for improved control of CINV.
- APF530 is an extended-release tri(ethylene glycol) poly(ortho ester) (TEG-POE)-based polymer formulation containing the 5-HT3 receptor antagonist granisetron and is designed to deliver granisetron over a 5-day period after a single subcutaneous (SC) injection in the abdomen.
- A large phase 3 trial was conducted to compare SC APF530 with intravenous (IV) palonosetron. Primary endpoints included noninferiority to palonosetron in preventing acute and delayed CINV after administration of moderately emetogenic chemotherapy (MEC), and noninferiority in preventing acute CINV after highly emetogenic chemotherapy (HEC). Positive results from this noninferiority study were previously presented.[1]
- Because assessment of emesis is an important component of best practice in clinical guidelines,[2-4] nausea severity and patient satisfaction with nausea control were specifically assessed as key secondary endpoints in this trial.
- A history of prior chemotherapy is one of several patient-related factors associated with increased risk for CINV.[2,3] Therefore, a subgroup analysis was performed on this vulnerable chemotherapy-nonnaïve subset of enrolled patients.

METHODOLOGY

Study design: Randomized, multicenter, observer-blind, double-dummy, parallel-group phase 3 study **(Figure 1)**

Participants: Chemotherapy-naïve or nonnaïve men or women ≥18 years old

Chemotherapy and study drug administration

- Patients received single-day treatment with MEC or HEC as defined by Hesketh[5] and were stratified at randomization by emetogenicity of chemotherapy.
 - For the purpose of the analysis performed here, patients receiving MEC and HEC were combined into a single population.
- APF530 (single SC injection in the abdomen) and palonosetron (IV infusion) or placebo were administered 30 to 60 minutes before chemotherapy.
 - Placebo for both SC and IV injections was isotonic saline.
- Standard doses of dexamethasone were administered with each study drug.
 - If MEC: Dexamethasone 8 mg IV on Day 1, then none on Days 2-4.
 - If HEC: Dexamethasone 20 mg IV on Day 1, then 8 mg PO BID on Days 2-4.

Figure 1. Study Design

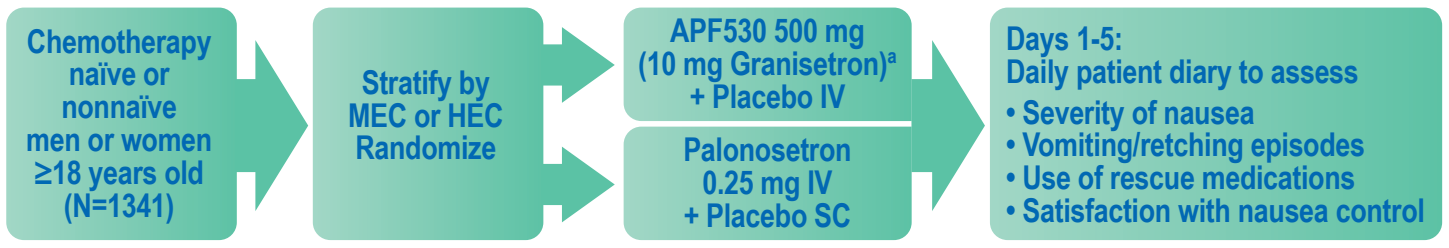
Chemotherapy naïve or nonnaïve men or women ≥18 years old (N=1341)
Stratify by MEC or HEC Randomize
APF530 500 mg (10 mg Granisetron)a + Placebo IV
Palonosetron 0.25 mg IV + Placebo SC
Days 1-5: Daily patient diary to assess
• Severity of nausea
• Vomiting/retching episodes
• Use of rescue medications
• Satisfaction with nausea control

[a]A third treatment group given APF530 250 mg (5 mg granisetron) + placebo IV was also studied and is not reported here.

OUTCOME MEASURES

- Complete response (CR) was defined as no emetic episodes and no use of rescue medication. Post-hoc subgroup analysis of data was performed for chemotherapy-naïve and nonnaïve patients.
- Patients used a daily diary to record severity of nausea, vomiting/retching episodes, use of rescue medication, and satisfaction with nausea/vomiting control.
- Assessments were made on each treatment day and for the overall 5-day treatment period. Nausea severity was graded as:
 - None
 - Mild (easily tolerated, did not interfere with normal daily activities)
 - Moderate (caused some interference with daily activities)
 - Severe (all normal activities completely stopped due to nausea)

 Satisfaction with overall control was scored as:
 - Very satisfied
 - Satisfied
 - Neither satisfied nor dissatisfied
 - Dissatisfied
 - Very dissatisfied

DEMOGRAPHICS

A total of 1341 randomized patients received study drug at 103 sites in the United States, India, and Poland and comprised the modified intent-to-treat (mITT) population. **Table 1** summarizes the demographics of patients in each chemotherapy stratum who were treated with 1 of 2 APF530 doses or palonosetron in Cycle 1. Breast cancer was the most common cancer type in the overall patient population (44%), followed by lung (18%) and ovarian (12%) cancers. Patients received chemotherapy regimens listed in **Table 2**.

Table 1. Demographics

Parameter	MEC		HEC	
	APF530	Palonosetron	APF530	Palonosetron
mITT[a] Population (n)	212	208	240	238
Mean Age ± SD (years)	55	57	57	58
Female (%)	84	83	63	67
Ethnicity (%)				
Caucasian	56	67	62	61
Asian	27	22	27	24
Prior Chemotherapy (%)	47	49	58	56

[a]Modified intent-to-treat.

Table 2. Chemotherapy Regimens

MEC[a] (% of Patients)		HEC[a] (% of Patients)	
Doxorubicin + Cyclophosphamide	54	Carboplatin combination	49
Cyclophosphamide mono- or combination	15	Cisplatin / Other combination	22
Carboplatin mono- or combination	11	Doxorubicin + Cyclophosphamide	15
Doxorubicin mono- or combination	6	Cyclophosphamide / Other combination	6
Irinotecan mono- or combination	5	Doxorubicin / Other combination	5
Other	9	Oxaliplatin combination	2
		Other	1

[a]Classified per Hesketh criteria.[5]

RESULTS

COMPLETE RESPONSE

- There was no statistical difference between APF530 and palonosetron in the rate of CR achieved by either chemotherapy-naïve and chemotherapy-nonnaïve individuals observed over the entire 120-hour period in the combined MEC and HEC population **(Figure 2)**.

Figure 2. CR – Chemotherapy-Nonnaïve vs. Naïve Patients in the 120-Hour Period

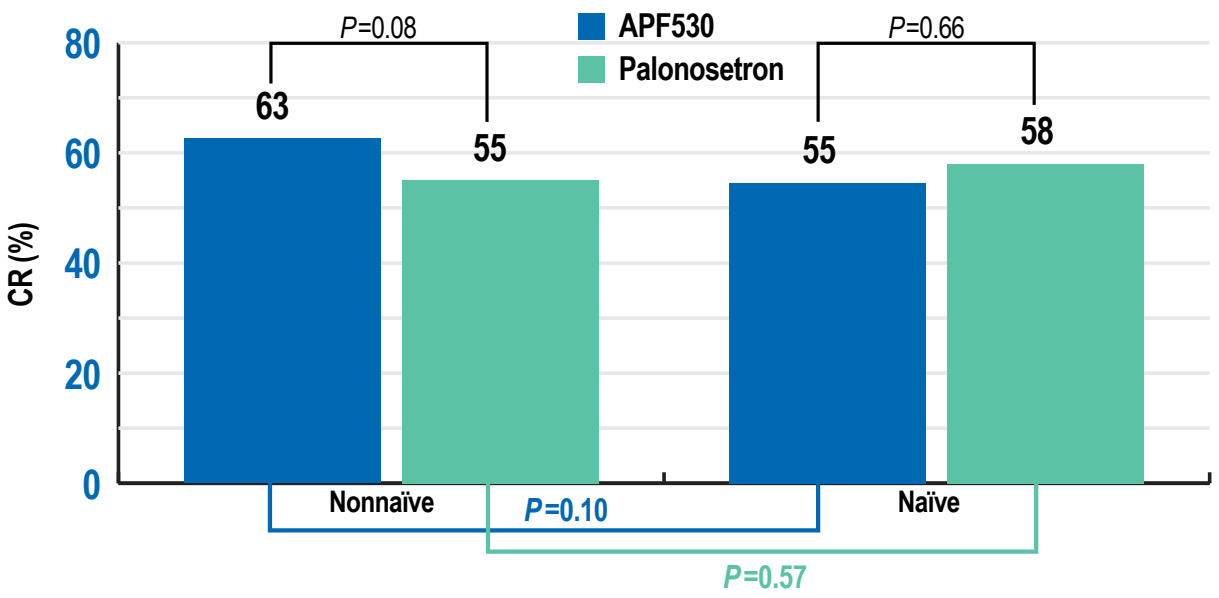
APF530
Palonosetron
CR (%)
0
20
40
60
80
P=0.08
P=0.66
63
55
55
58
Nonnaïve
Naïve
P=0.10
P=0.57

NAUSEA CONTROL

- There was no statistical difference between APF530 and palonosetron in nausea severity in either chemotherapy-naïve or chemotherapy-nonnaïve individuals in the combined MEC and HEC population.
- For each day, there were no statistical differences in nausea severity in either arm, in patient subgroups, or in the entire population at an experimental error rate of 0.05 **(Figure 3)**.
- For the entire 5-day period, 69% to 71% of all patients reported a maximum nausea severity of "None" or "Mild" with either treatment (no statistical difference).
- On Day 5, moderate-to-severe nausea was reduced to 13% in APF530 patients and 10% in palonosetron patients regardless of previous exposure to chemotherapy (no statistical difference).

Figure 3. Nausea Severity

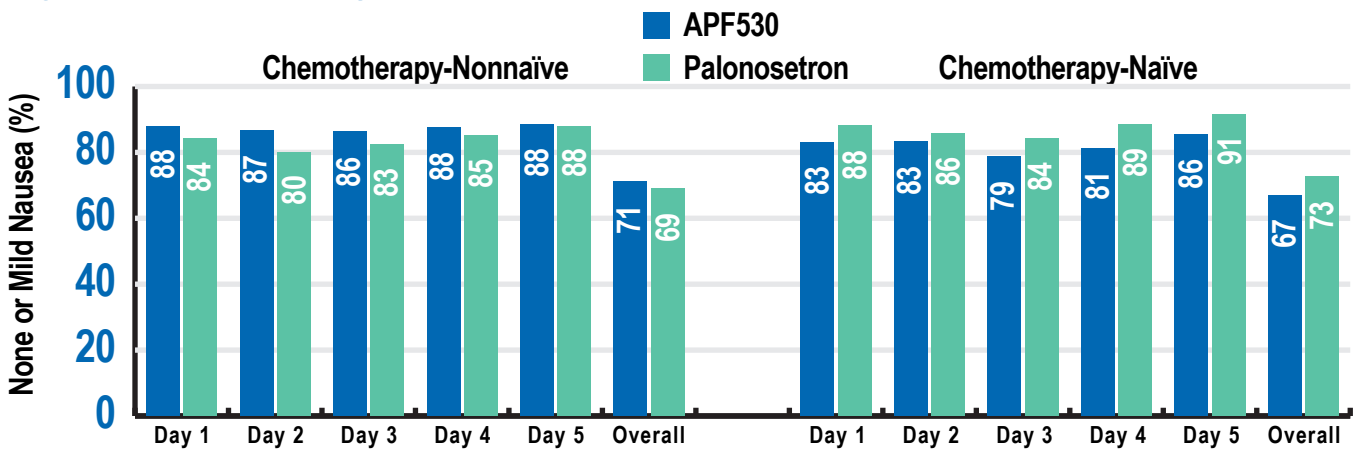
APF530
Palonosetron
Chemotherapy-Nonnaïve
Chemotherapy-Naïve
None or Mild Nausea (%)
0
20
40
60
80
100
88 84
87 80
86 83
88 85
88 88
71 69
83 88
83 86
79 84
81 89
86 91
67 73
Day 1
Day 2
Day 3
Day 4
Day 5
Overall
Day 1
Day 2
Day 3
Day 4
Day 5
Overall

PATIENT SATISFACTION

- There was no statistical difference between APF530 and palonosetron in patient satisfaction in either chemotherapy-naïve or chemotherapy-nonnaïve individuals in the combined MEC and HEC population.
- For each day, there were no statistical differences in patient satisfaction in either arm, in patient subgroups, or in the entire population at an experimental error rate of 0.05 **(Figure 4)**.
- For the entire 5-day period, similar proportions of all patients (70% of APF530 patients and 75% of palonosetron patients) reported their worst assessment of nausea control as either "Very Satisfied" or "Satisfied" (no statistical difference).

Figure 4. Patient Satisfaction

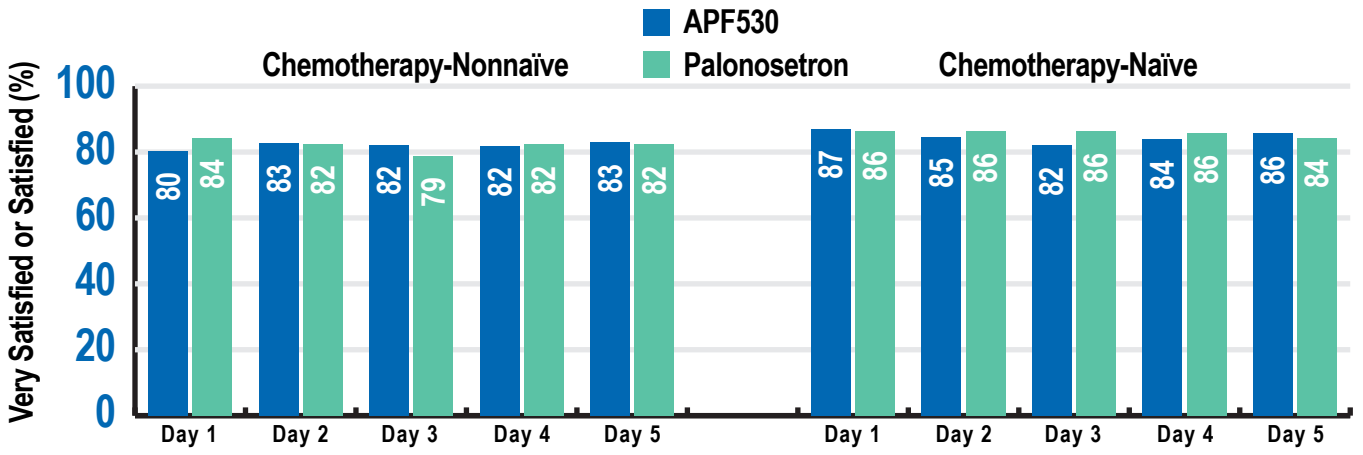
APF530
Palonosetron
Chemotherapy-Nonnaïve
Chemotherapy-Naïve
Very Satisfied or Satisfied (%)
0
20
40
60
80
100
80 84
83 82
82 79
82 82
83 82
87 86
85 86
82 86
84 86
86 84
Day 1
Day 2
Day 3
Day 4
Day 5
Day 1
Day 2
Day 3
Day 4
Day 5

CONCLUSIONS

- APF530 provides comparable CR rates to palonosetron over the 120-hour period whether or not the patient had previous exposure to chemotherapy.
- Severity of nausea and patient satisfaction were similar with APF530 and palonosetron after administration of MEC or HEC, regardless of previous chemotherapy exposure.
- APF530 offers comparable nausea control and patient satisfaction to palonosetron over a 5-day period.

References: 1. Grous JJ, Gabrail N, Charu V, et al. Phase 3 study of sustained release granisetron (APF530) compared to palonosetron for the prevention of chemotherapy-induced nausea and vomiting. *J Clin Oncol.* 27:15s, 2009(suppl; abstr 9627); presented at the 45th American Society of Clinical Investigation Annual Meeting, Orlando, FL, 2009. **2.** Roila F, Herrstedt J, Aapro M, et al; the ESMO/MASCC Guidelines Working Group. Guideline update for MASCC and ESMO in the prevention of chemotherapy- and radiotherapy-induced nausea and vomiting: results of the Perugia consensus conference. *Ann Oncol.* 2012;21(suppl 5):v232-v243. **3.** National Comprehensive Cancer Network. NCCN clinical practice guidelines in oncology: antiemesis. Version 1.2012. **4.** Basch E, Prestrud AA, Hesketh PJ, et al. Antiemetics: American Society of Clinical Oncology Clinical Practice Guideline Update. *J Clin Oncol.* 2011;29(31):4189-4198. **5.** Hesketh PJ. Defining the emetogenicity of cancer chemotherapy regimens: relevance to clinical practice. *Oncologist.* 1999;4(3):191-196.

Acknowledgements:
Financial support: A.P. Pharma, Redwood City, CA, USA.
Editorial assistance: Gemini Healthcare, LLC, Westbrook, CT, USA.

Abstract # 1109