4-143

Opioid-free Postoperative Recovery in Patients Undergoing Herniorrhaphy With HTX-011 as the Foundation of a Scheduled, Non-opioid Multimodal Analgesic Regimen

Jackie Evans-Shields,[1] Eugene R. Viscusi,[2] Harold Minkowitz,[3] Peter Winkle,[4] Jia Hu,[1] Neil Singla[5]

[1]Heron Therapeutics, Inc., San Diego, CA, USA; [2]Sidney Kimmel Medical College of Thomas Jefferson University, Philadelphia, PA, USA; [3]HD Research, LLC, Houston, TX, USA; [4]Anaheim Clinical Trials, Anaheim, CA, USA; [5]Lotus Clinical Research, LLC, Pasadena, CA, USA

INTRODUCTION

- Most severe pain occurs within the first 72 hours after surgery[1,2]
- Opioid medications are commonly prescribed following surgery
 - Opioids can be associated with significant side effects and they have the potential to lead to long-term use and dependence[3-5]
- HTX-011 is an extended-release dual-acting local anesthetic combining bupivacaine and low-dose meloxicam in a proprietary Biochronomer™ polymer that allows for the diffusion of active ingredients over 72 hours (Heron Therapeutics, Inc., San Diego, CA)
 - The combination of bupivacaine with meloxicam reduces inflammation and acidity at the surgical site, allowing increased penetration of bupivacaine into neurons (**Figure 1**)
 - HTX-011 is administered via needle-free application to the surgical site prior to closure using a syringe and a Luer-lock applicator (**Figure 2**)

Figure 1. HTX-011 Dual Mechanism of Action

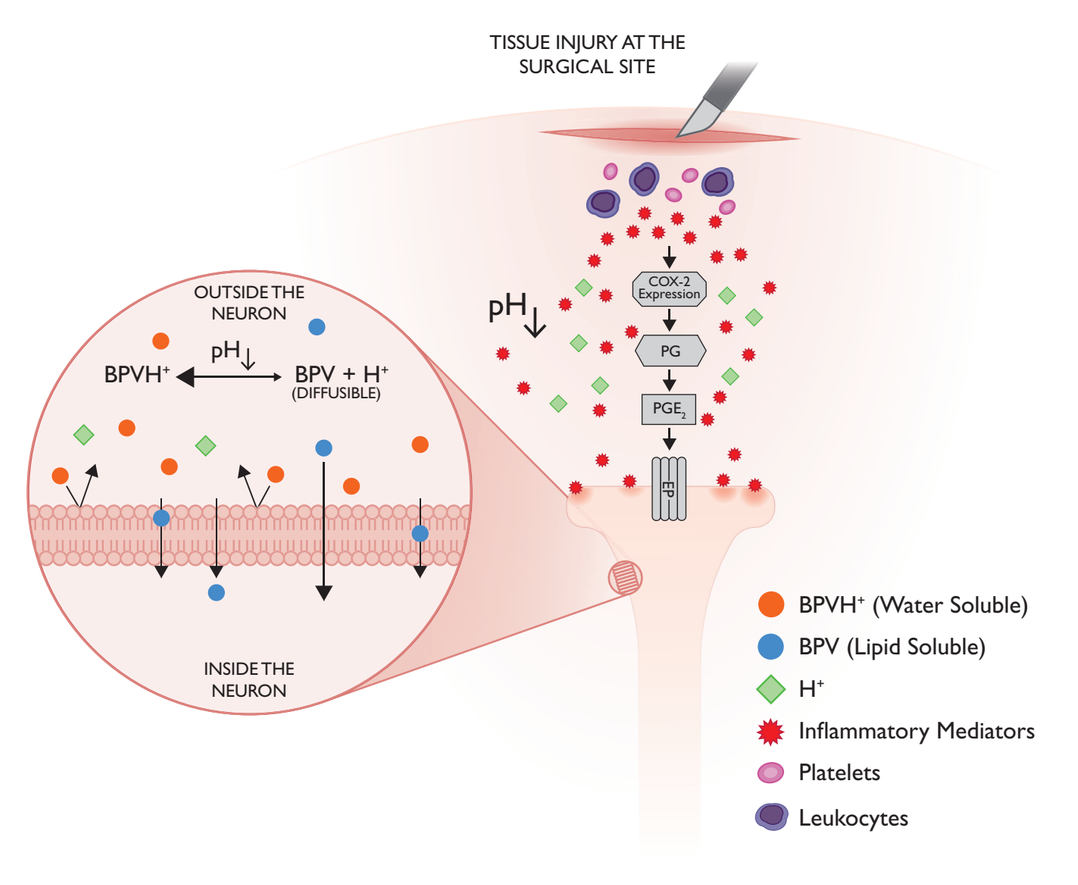
TISSUE INJURY AT THE SURGICAL SITE
OUTSIDE THE NEURON
pH↓
BPVH⁺
BPV + H⁺ (DIFFUSIBLE)
COX-2 Expression
PG
PGE₂
INSIDE THE NEURON
BPVH⁺ (Water Soluble)
BPV (Lipid Soluble)
H⁺
Inflammatory Mediators
Platelets
Leukocytes

Figure 2. Needle-Free Administration of HTX-011

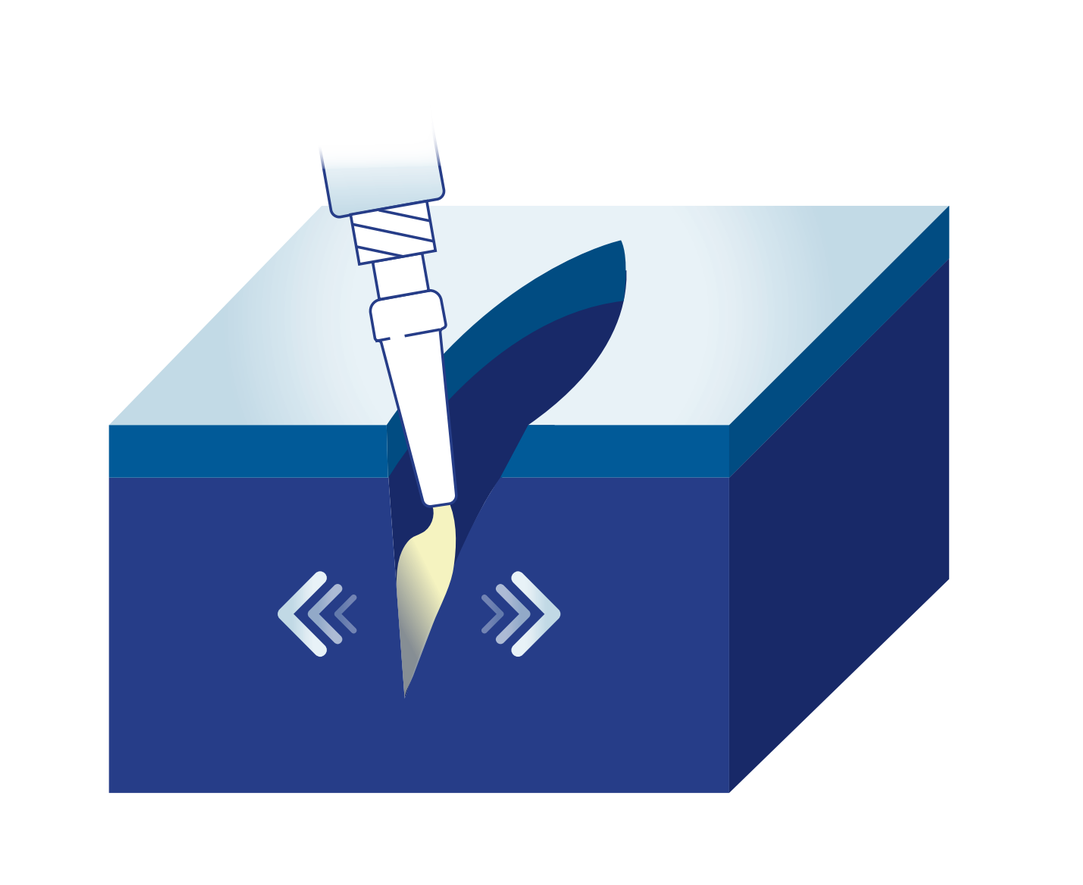

- In a prior phase 3 herniorrhaphy study (EPOCH-2; NCT03237481)[6] comparing HTX-011 300 mg/9 mg (bupivacaine/meloxicam) vs bupivacaine HCl 75 mg and saline placebo, HTX-011:
 - Provided superior pain relief over the first 72 hours
 - Significantly reduced incidence of severe pain at any time over the first 72 hours
 - Significantly reduced total opioid consumption through 72 hours following surgery
 - Allowed 95% of patients who were opioid-free during the 72-hour postoperative period to remain opioid-free through Day 10
- As most patients are discharged on the same day following herniorrhaphy,[7] the ability to rapidly predict which patients will not require opioids has the potential to dramatically reduce unnecessary opioid prescriptions, while still maintaining effective analgesia following surgery

OBJECTIVES

- The objectives of this follow-on study were to:
 - Assess the proportion of patients remaining opioid-free with HTX-011 plus a scheduled non-opioid MMA regimen during the first 72 hours following herniorrhaphy
 - Assess total opioid use in this population
 - Assess the proportion of patients who were opioid-free during the first 72 hours and remained opioid-free through Day 10 and Day 28
 - Assess the relationship between opioid use and severe pain

METHODS

Study Design

- The EPOCH 2 Follow-on Study enrolled patients undergoing open inguinal herniorrhaphy with mesh who met study criteria (**Table 1**)
 - As the superiority of HTX-011 to bupivacaine HCl was previously demonstrated in a phase 3 study in the same treatment population,[6] an active control was not included in this study
- All patients (Cohorts 1 and 2) received a single dose of HTX-011 300 mg/9 mg (bupivacaine/meloxicam) administered via needle-free application to the surgical site prior to closure in addition to non-opioid MMA therapy consisting of:
 - Preoperative oral acetaminophen 1 g
 - Postoperative oral ibuprofen 600 mg and oral acetaminophen 1 g every 6 hours, alternating, so that patients received an analgesic every 3 hours
 - Cohort 2 also received a single dose of intravenous (IV) ketorolac intraoperatively (15-30 mg, per prescribing information)[8]
- Following surgery, patients remained in the hospital for 72 hours; upon discharge, patients were provided a daily diary to record whether they took an opioid medication between 72 hours and Day 28
 - Patients were instructed to continue their MMA regimen as needed (ibuprofen for initial rescue supplemented with acetaminophen if pain persisted)
- Opioid rescue medication was available only upon request during the first 72 hours, independent of pain score
 - Patients who received <10 mg immediate-release oxycodone by mouth within 12 hours before discharge were not to be provided an opioid prescription

Table 1. Key Inclusion and Exclusion Criteria

Key Inclusion Criteria	Key Exclusion Criteria
• Adult males and females who are not pregnant or lactating • Scheduled to undergo unilateral open inguinal herniorrhaphy with mesh under general anesthesia • ASA Physical Status Classification System category 1-3	• Pre-existing acute or chronic painful/restrictive condition that may require analgesia during the postoperative period • Use of the following within a defined period prior to surgery: – NSAIDs within 10 days – Bupivacaine within 5 days – Long-acting opioids within 3 days – Any opioid within 24 hours – Any local anesthetic within 24 hours • BMI >39 kg/m²

ASA, American Society of Anesthesiologists; BMI, body mass index; NSAID, nonsteroidal anti-inflammatory drugs.

Outcome Measures

- The primary endpoint was the proportion of patients who remained opioid-free through 72 hours following surgery
- Additional endpoints included:
 - Proportion of patients who experienced severe pain at any time through 72 hours
 - Total opioid use through 72 hours
 - Proportion of patients receiving no opioid rescue through 72 hours who remained opioid-free through the 28-day recovery period
 - Incidence of adverse events (AEs), serious AEs, and opioid-related AEs

Assessments

- Opioid use was monitored by site staff through 72 hours following surgery and from discharge through study end using a patient-maintained daily opioid diary for recording opioid use (yes/no)
 - During the 72-hour inpatient period, patients were only considered opioid-free if total postoperative opioid morphine milligram equivalents (MME) administered was 0
 - From 72 hours through Day 28, patients were only considered opioid-free if all diary entries were completed with the response "no"; patients with missing or incomplete diary entries were not considered to be opioid-free
- Postoperative pain was assessed using a numeric rating scale (NRS), where 0 indicates "no pain" and 10 indicates "worst pain imaginable"
 - To reduce the confounding effects of opioids, pain intensity scores during periods of opioid administration were replaced with the highest observed scores before opioid medication use

RESULTS

Disposition and Baseline Characteristics

- Sixty-three patients were treated with HTX-011 across 3 study sites; 58 (92%) completed the study through Day 28
- Most patients (94%) were male (consistent with the general hernia population) with a mean (SD) age of 49 (14) years and a mean (SD) body mass index of 28 (4) kg/m²

Postoperative Opioid Use

- In this follow-on study, 90.5% of patients (57/63) receiving HTX-011 with a non-opioid MMA regimen remained opioid-free through 72 hours following surgery, and 91.2% of them (52/57) remained opioid-free through Day 28 of recovery; there was no significant benefit with the addition of ketorolac and the cohorts were therefore combined (**Figure 3**)
 - In comparison, 51% of patients (84/164) in EPOCH-2 receiving HTX-011 alone remained opioid-free through 72 hours, and 84.5% of them (71/84) remained opioid free through Day 28[6]

Figure 3. 91% of Herniorrhaphy Patients Remained Opioid-free Through 72 Hours With HTX-011 Plus Scheduled Non-Opioid MMA

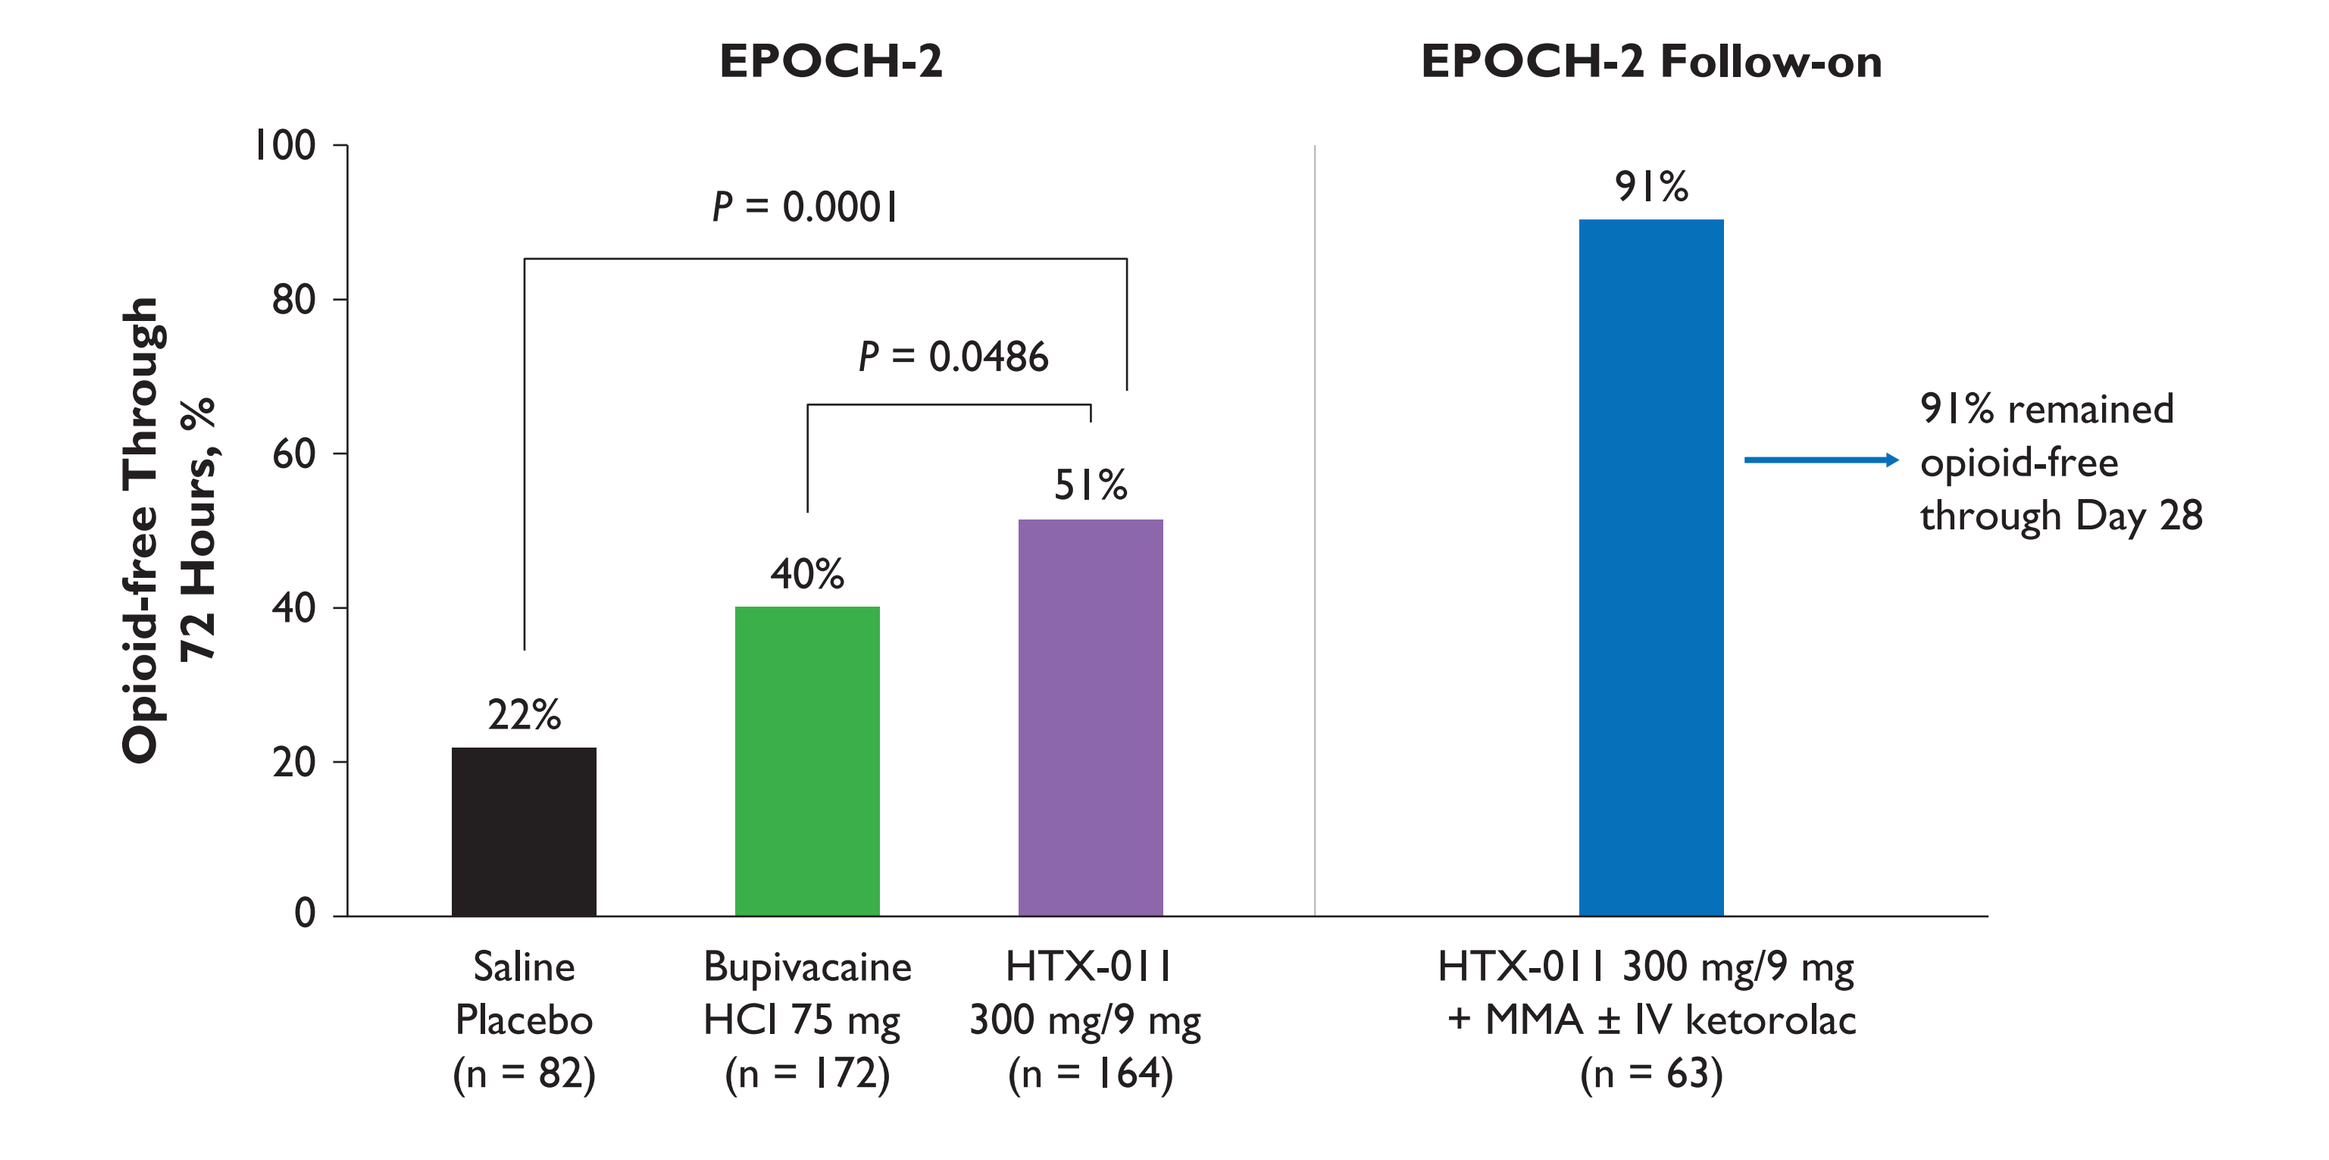
EPOCH-2
EPOCH-2 Follow-on
Opioid-free Through 72 Hours, %
100
80
60
40
20
0
P = 0.0001
P = 0.0486
22%
40%
51%
91%
91% remained opioid-free through Day 28
Saline Placebo (n = 82)
Bupivacaine HCl 75 mg (n = 172)
HTX-011 300 mg/9 mg (n = 164)
HTX-011 300 mg/9 mg + MMA ± IV ketorolac (n = 63)

HCl, hydrochloride; IV, intravenous; MMA, multimodal analgesia.

- Mean total opioid consumption (in MME) was significantly reduced in patients treated with HTX-011 and a scheduled, non-opioid MMA regimen in this follow-on study (0.94) compared with those receiving saline placebo in EPOCH-2 (17.53)[6] (**Table 2**)

Table 2. Total Postoperative Opioid Consumption (MME) Through 72 Hours

	EPOCH-2 Study			EPOCH-2 Follow-on Study[a]
	HTX-011 300 mg/9 mg n = 164	Bupivacaine HCl 75 mg n = 172	Saline Placebo n = 82	HTX-011 + MMA n = 63
Total postoperative opioid consumption through 72 hours				
Mean (SD)	10.85 (17.062)	14.51 (18.185)	17.53 (18.908)	0.94 (3.278)
Median (Min, Max)	0.00 (0.0, 103.0)	7.25 (0.0, 87.5)	11.25 (0.0, 73.5)	0.00 (0.0, 18.5)

HCl, hydrochloride; max, maximum, min, minimum; MMA, multimodal analgesia; MME, morphine milligram equivalents; SD, standard deviation.
[a]Both cohorts combined; opioid consumption was similar between cohorts.

Pain Intensity

- Mean pain intensity was mild (NRS <4) at every assessment timepoint during the 72-hour inpatient period; ketorolac did not confer additional benefit (**Figure 4**)
- In total, 82.5% of patients (52/63) did not experience severe pain (NRS ≥7) at any time following surgery
 - Among patients who experienced severe pain (11/63), all initially reported it within the first 24 hours following surgery

Figure 4. HTX-011 With a Non-Opioid MMA Effectively Controlled Postoperative Pain

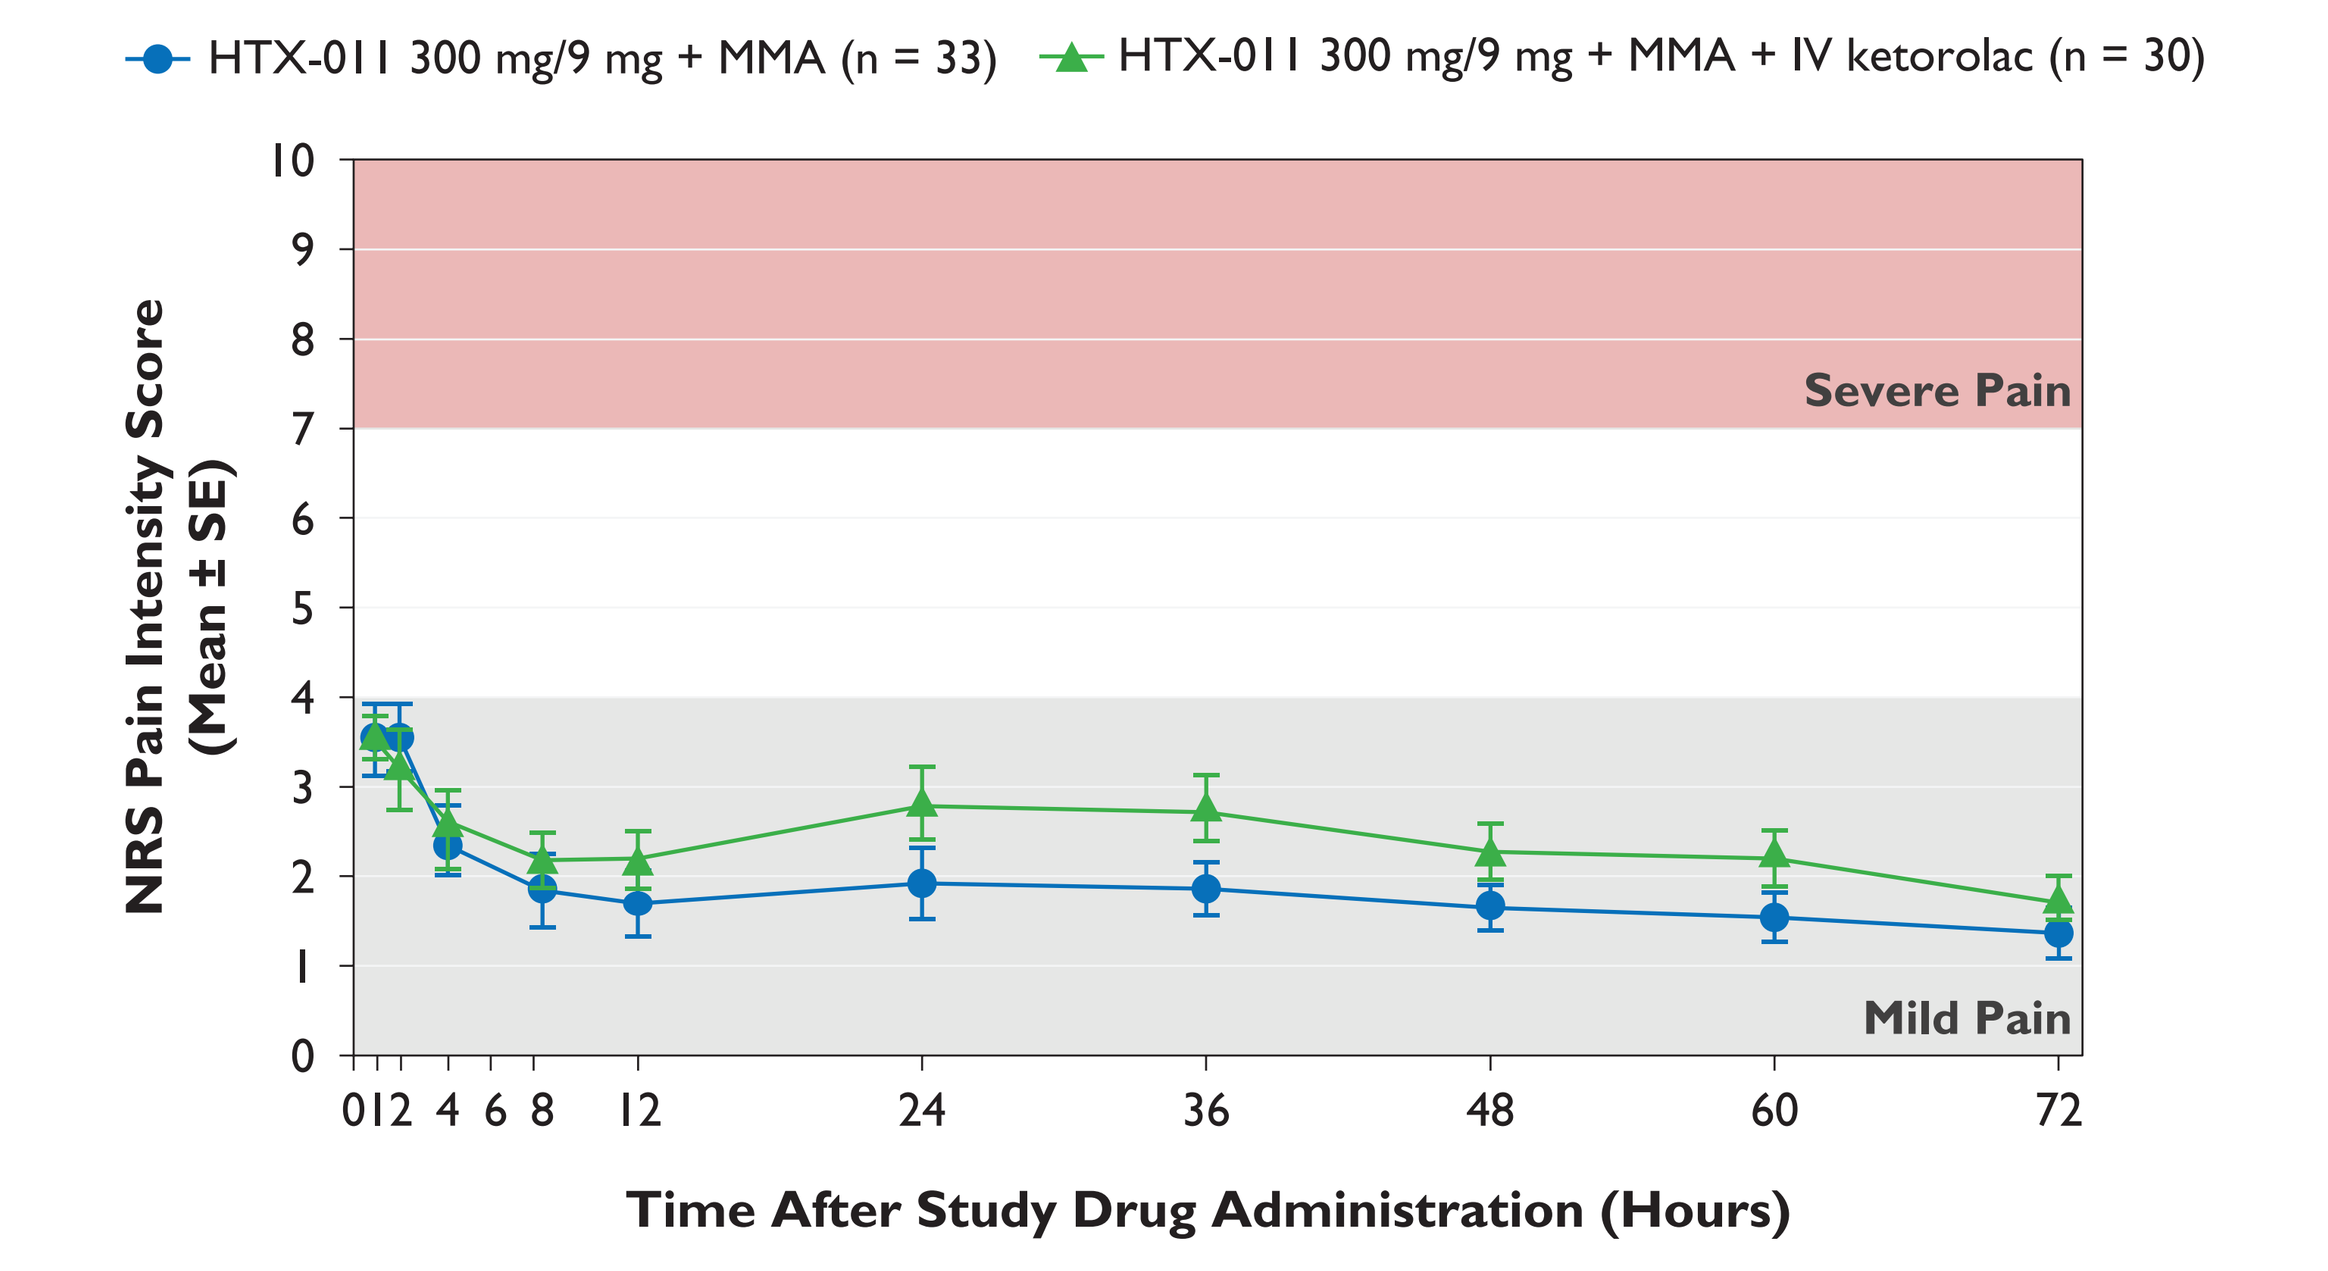
HTX-011 300 mg/9 mg + MMA (n = 33)
HTX-011 300 mg/9 mg + MMA + IV ketorolac (n = 30)
NRS Pain Intensity Score (Mean ± SE)
10
9
8
7
6
5
4
3
2
1
0
Severe Pain
Mild Pain
0 1 2 4 6 8 12 24 36 48 60 72
Time After Study Drug Administration (Hours)

IV, intravenous; MMA, multimodal analgesia; NRS, numeric rating scale; SE, standard error.

Relationship Between Opioid Use and Pain Intensity

- The 6 patients requiring opioid rescue exhibited a distinct pain profile within the first 2 hours following surgery, which was maintained throughout the 72-hour postoperative period (**Figure 5**)

Figure 5. Patients Requiring Opioids Can be Identified Early[a]

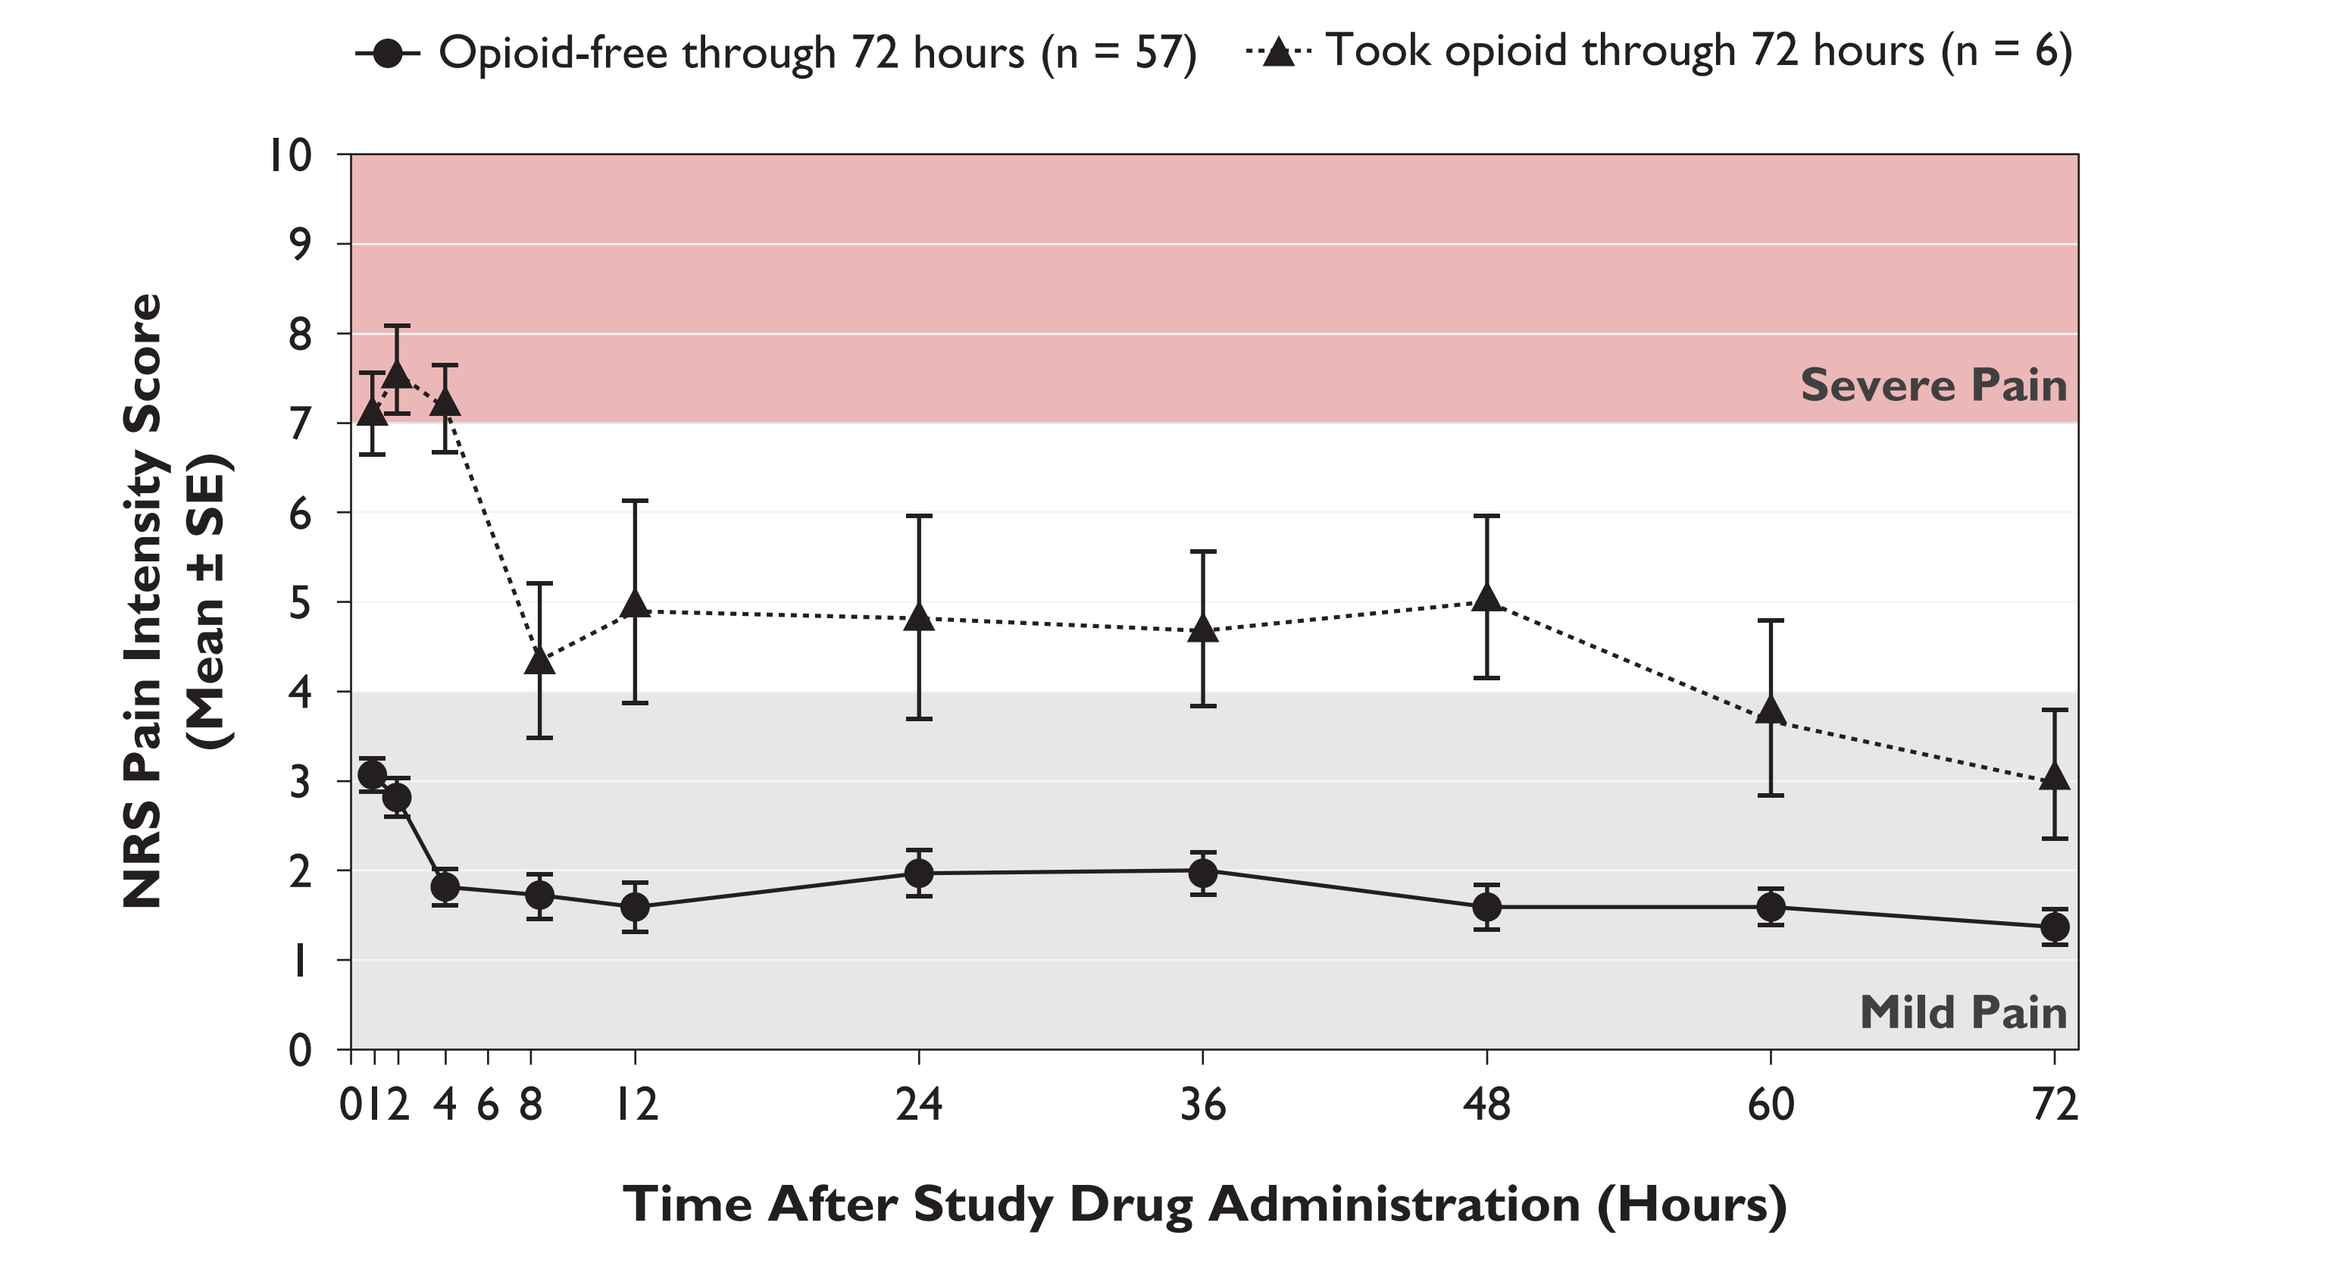
Opioid-free through 72 hours (n = 57)
Took opioid through 72 hours (n = 6)
NRS Pain Intensity Score (Mean ± SE)
10
9
8
7
6
5
4
3
2
1
0
Severe Pain
Mild Pain
0 1 2 4 6 8 12 24 36 48 60 72
Time After Study Drug Administration (Hours)

NRS, numeric rating scale; SE, standard error.
[a]Combined data from both cohorts.

- All 6 patients (10%) requesting an opioid medication during the 72-hour postoperative period reported an NRS score of ≥6 within 1 hour following surgery; 4 of these patients reported severe pain (NRS ≥7) within the first 2 hours following surgery
 - All patients who took an opioid during the 72-hour inpatient period reported severe pain and/or received an opioid within 2 hours following surgery

Safety

- Thirty-eight percent of patients (24/63) experienced an AE; none of these were serious or led to study withdrawal (**Table 3**)
 - The incidence of opioid-related AEs in this study was lower than the prior phase 3 study, concordant with the decreased use of opioids[6]
- There was no evidence of nonsteroidal anti-inflammatory drug (NSAID)-related toxicity with the addition of an MMA regimen (which included NSAIDs) to HTX-011

Table 3. Summary of Adverse Events, n (%)

Category	EPOCH-2 Study			EPOCH-2 Follow-on Study[a]
	HTX-011 n = 163	Bupivacaine HCl n = 173	Saline Placebo n = 82	HTX-011 + MMA n = 63
Any AE	119 (73.0)	127 (73.4)	61 (74.4)	24 (38.1)
Opioid-related AE	53 (32.5)	73 (42.2)	36 (43.9)	7 (11.1)[b]
AE leading to study discontinuation	0	0	0	0
SAE	2 (1.2)	1 (0.6)	1 (1.2)	0
Fatal SAE	0	0	0	0

AE, adverse event; HCl, hydrochloride; IV, intravenous; MMA, multimodal analgesia; SAE, serious adverse event.
[a]Both cohorts combined; rates of AEs were similar between cohorts.
[b]Opioid-related AEs observed in the follow-on study included constipation, nausea, pruritis, and vomiting.

CONCLUSIONS

- 91% of patients were opioid-free when HTX-011 was used as the foundation of a scheduled non-opioid MMA regimen following hernia repair
 - The mean pain intensity never rose above the mild range
 - Patients who did require an opioid exhibited a distinct pain profile from the first assessment at 1 hour following surgery
- More than 85% of patients never experienced severe postoperative pain
- No NSAID-related toxicity was observed, indicating the safety of combining HTX-011 with NSAID-containing MMA regimens
- IV ketorolac provided no additional benefit over HTX-011 + OTC MMA
- The following easy-to-use algorithm was formulated to assist in determining which patients should receive a prescription for opioids at discharge:
 - Only patients who receive an opioid and/or exhibit an NRS (0 to 10) pain score of ≥6 within the first 2 hours following surgery should receive a discharge prescription for opioids
 - This algorithm is being evaluated prospectively in the Helping Opioid Prescription Elimination (HOPE) Program (see ASHP Midyear 2019 Poster 4-144)

REFERENCES

1. Lynch EP, et al. *Anesth Analg*. 1997;85:117-123.
2. Svensson I, et al. *J Pain Symptom Manage*. 2000;20:193-201.
3. Hill MV, et al. *Ann Surg*. 2017;265:709-714.
4. Gan TJ. *J Pain Res*. 2017; 10:2287-2298.
5. Kessler ER, et al. *Pharmacotherapy*. 2013; 33:207-213.
6. Viscusi E, et al. *Hernia*. 2019. doi: 10.1007/s10029-019-02023-6. Published online August 19, 2019.
7. National Health System. Inguinal hernia repair. https://www.nhs.uk/conditions/inguinal-hernia-repair/what-happens/. Accessed October 21, 2019.
8. Ketorolac tromethamine [package insert]. Lake Forest, IL: Hospira, Inc; 2011.

ACKNOWLEDGMENTS

Funding for this research was provided by Heron Therapeutics, Inc. (San Diego, CA, USA). Medical writing assistance was provided by ApotheCom (San Francisco, CA, USA).

An electronic version of the poster can be viewed by scanning the QR code. The QR code is intended to provide scientific information for individual reference. The PDF should not be altered or reproduced in any way. All copyrights remain those of the copyright holder. This page will not be available after December 12, 2019. http://bit.ly/2N8fiBs **(This URL is case sensitive.)**

HERON THERAPEUTICS

Presented at the American Society of Health-System Pharmacists (ASHP) Midyear 2019 Congress; December 8-12, 2019; Las Vegas, NV, USA